M-Protein Analysis in Plasma of Multiple Myeloma Patients by Intact Mass Spectrometry

Luca Genovesi, Michael Schirm, Nick Dupuis, Gwenael Pottiez

CellCarta, Montreal, Quebec, Canada

POSTER NUMBER: DAY3-P24

CellCarta

INTRODUCTION

- Multiple myeloma (MM) is a type of cancer of the bone marrow characterized by an anormal growth of the number of plasma cells, which produce a monoclonal antibody (M-protein). Patients with MM have a moderate life expectancy and often require multiple lines of chemotherapy for durable control and long-term survival.[1]
- Measurable residual disease (MRD) is an important prognostic factor in MM to evaluate the depth of treatment response which is widely used in clinical trials. MRD is therefore monitored using techniques that measure the number of tumor cells in the bone marrow (BM).
- Multiparametric Flow Cytometry (MFC), Next Generation Sequencing (NGS), and imaging techniques, have been developed to provide sensitive MRD assessment, however, BM-MRD, is invasive and may not accurately represent disease burden due to BM site heterogeneity.[2]
- To overcome all these difficulties, Mass Spectrometry (MS) could offer non-invasive means of assessing the efficacy of the treatment, by measuring the M-protein in peripheral blood, bypassing the need for BM aspiration.[3]
- *Derman et al.*, showed that LC-MS out performs NGS and MALDI-TOF-MS in predicting PFS. LC-MS-negative patients showed no progression or death, indicating a higher sensitivity compared to the standard techniques. Data also showed that MS was able to detect relapse in earlier timepoints compared to NGS.[3]
- In this study, we present a blood-based method using mass spectrometry for M-protein detection and their applications of MRD detection in MM patients from assay characterization, to data analysis from plasma/serum samples in a clinical trial.

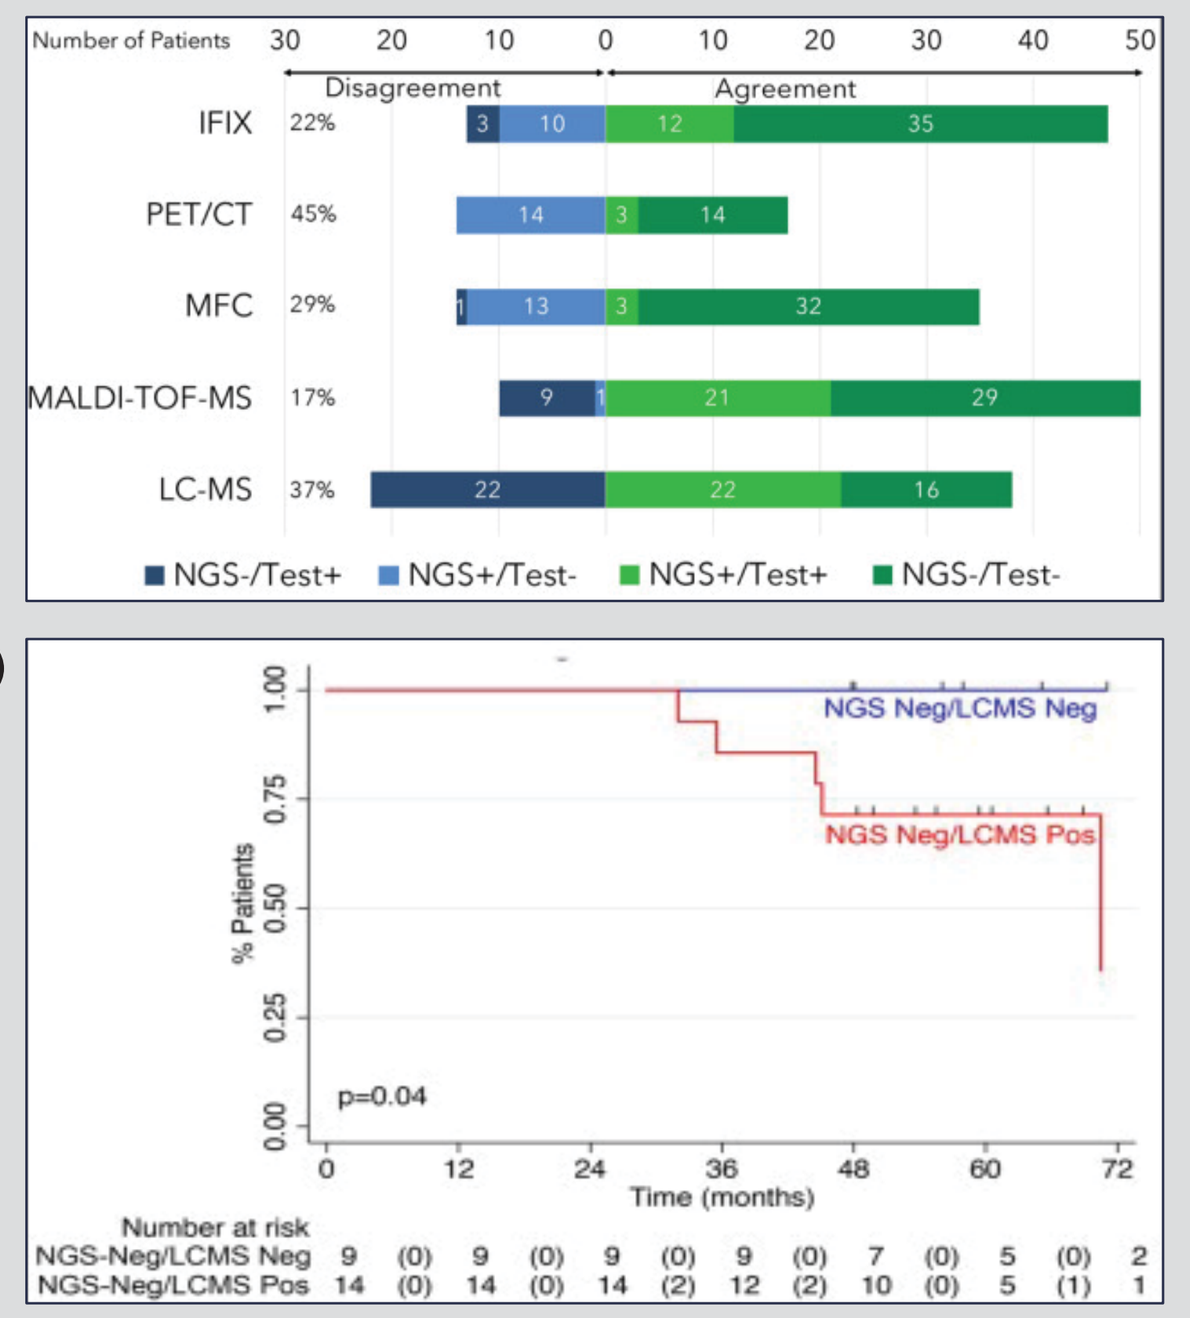

A)
Number of Patients
Disagreement
Agreement
IFIX 22%
PET/CT 45%
MFC 29%
MALDI-TOF-MS 17%
LC-MS 37%
NGS-/Test+
NGS+/Test-
NGS+/Test+
NGS-/Test-
B)
NGS Neg/LCMS Neg
NGS Neg/LCMS Pos
% Patients
p=0.04
Time (months)
Number at risk

Figure 1. A) Dis/agreement between NGS and other MRD assays. **B)** Progression-free survival (PFS) of all NGS–patients, stratified by LC-MS status.[3]

SAMPLE PROCESSING & DATA ANALYSIS WORKFLOW

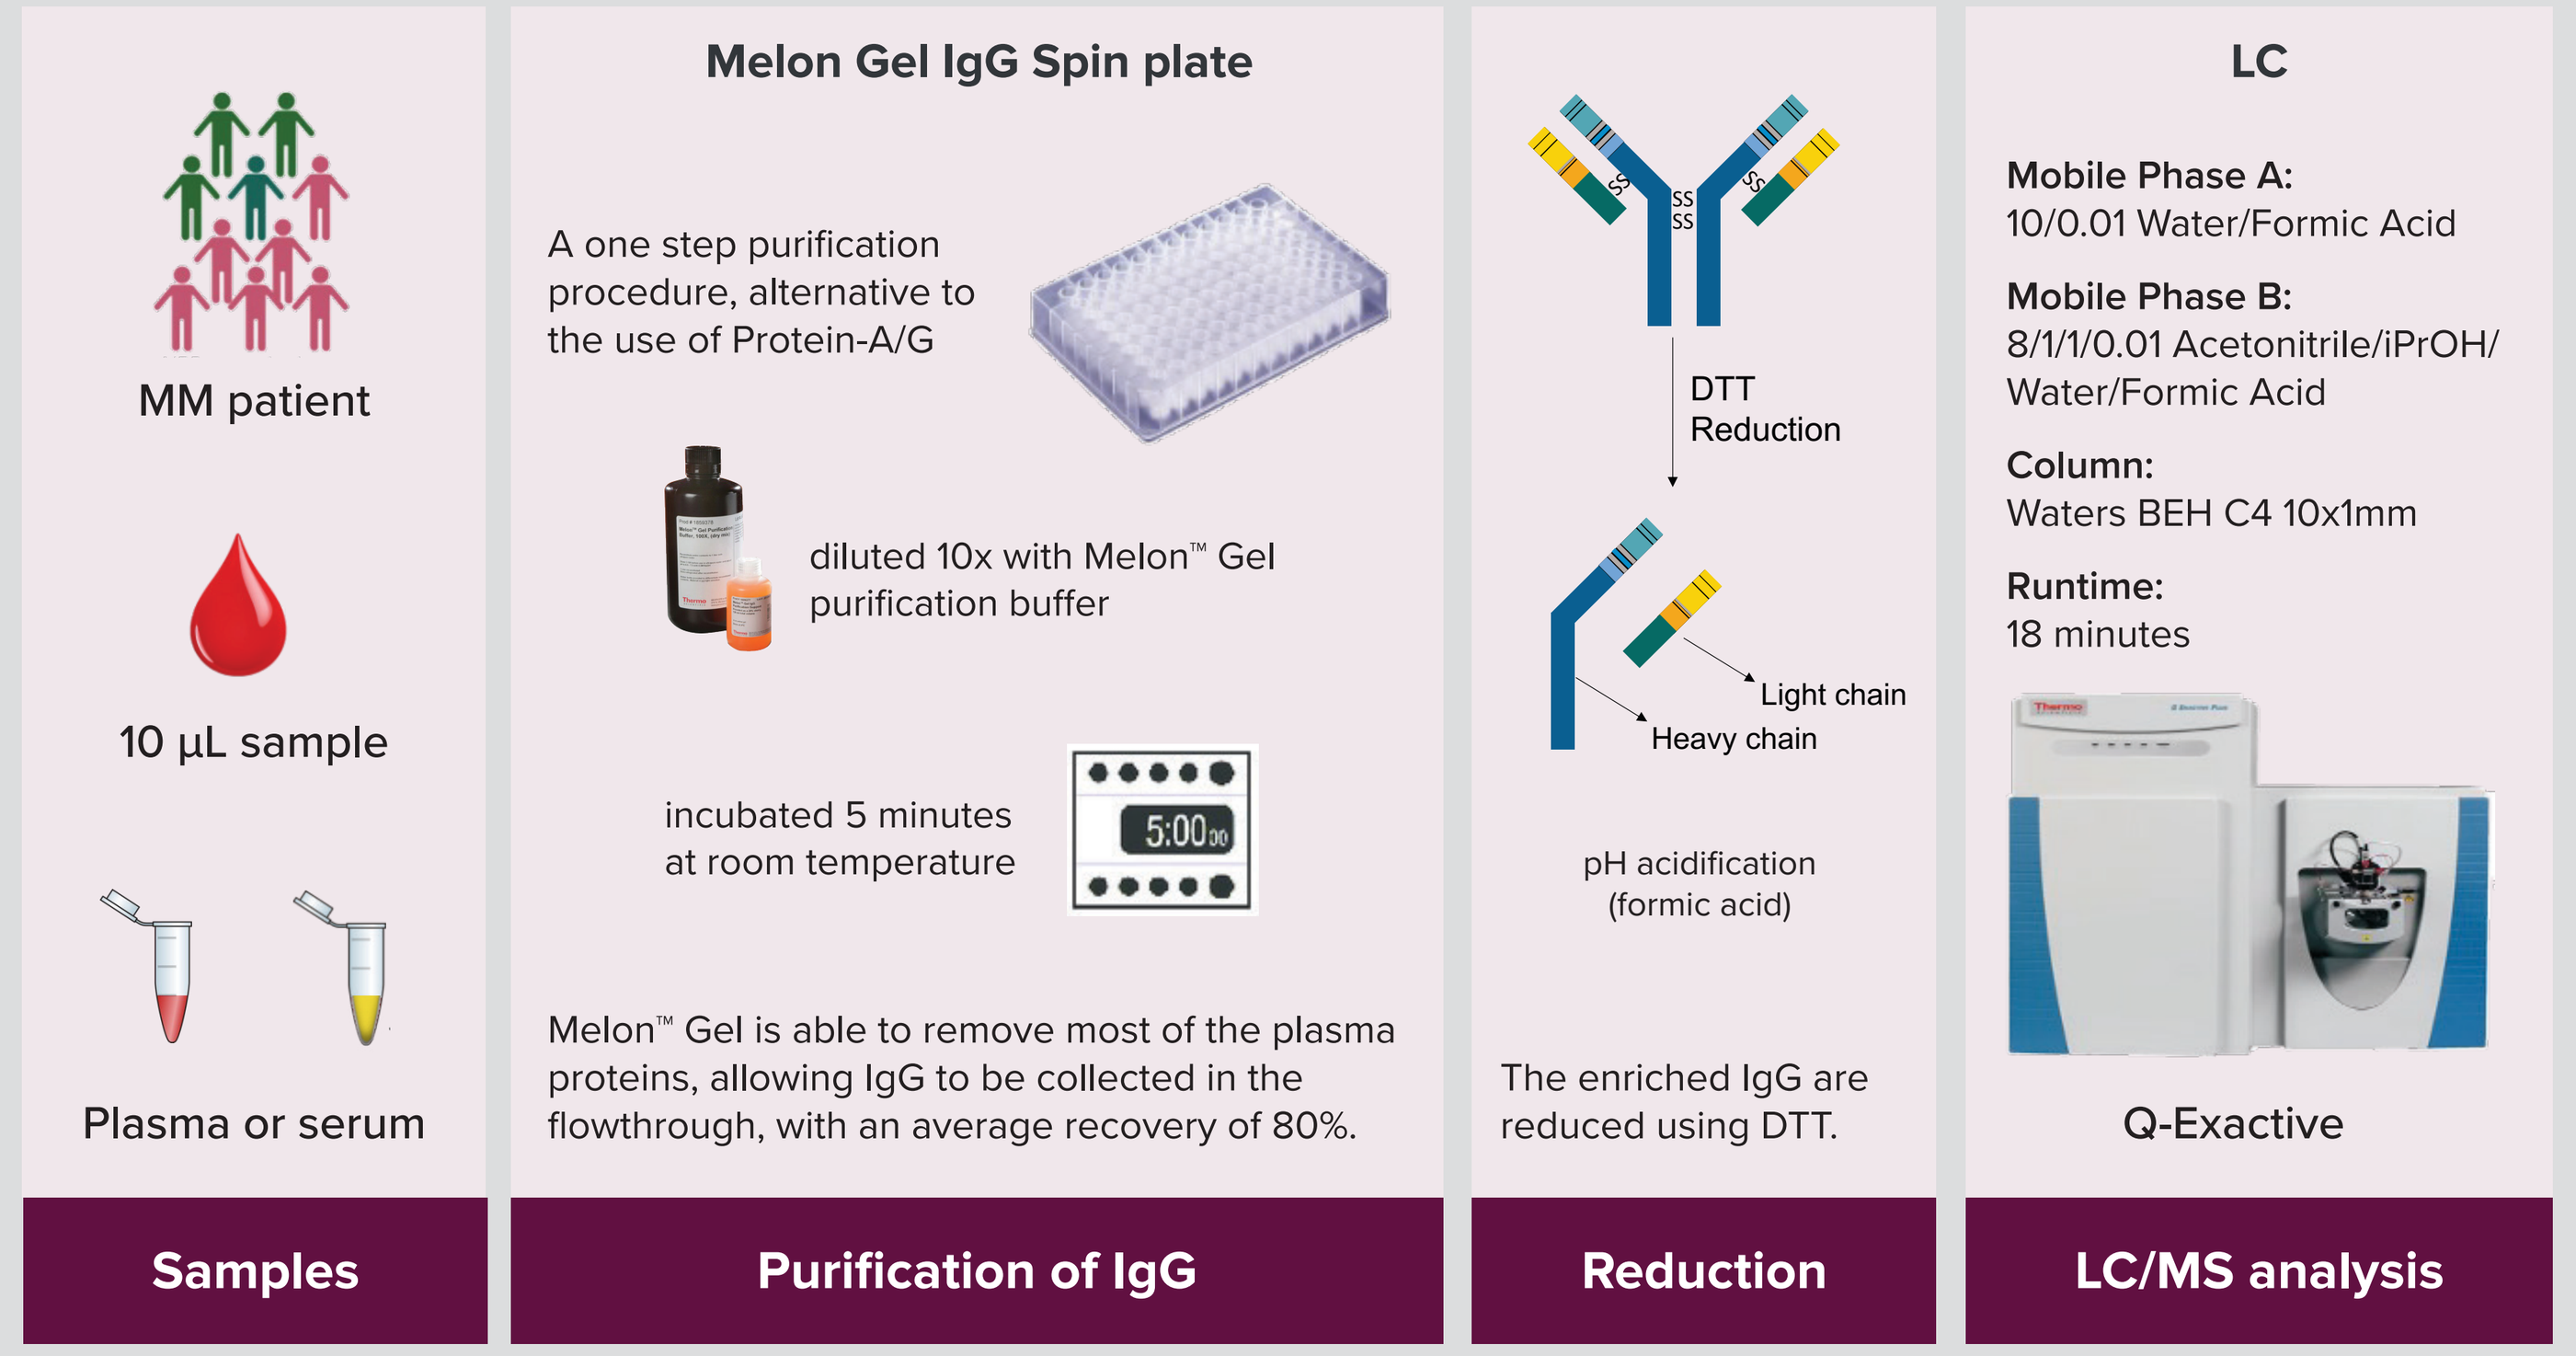

MM patient
10 µL sample
Plasma or serum
Samples
Melon Gel IgG Spin plate
A one step purification procedure, alternative to the use of Protein-A/G
diluted 10x with Melon™ Gel purification buffer
incubated 5 minutes at room temperature
5:00
Melon™ Gel is able to remove most of the plasma proteins, allowing IgG to be collected in the flowthrough, with an average recovery of 80%.
Purification of IgG
DTT Reduction
Light chain
Heavy chain
pH acidification (formic acid)
The enriched IgG are reduced using DTT.
Reduction
LC
Mobile Phase A: 10/0.01 Water/Formic Acid
Mobile Phase B: 8/1/1/0.01 Acetonitrile/iPrOH/ Water/Formic Acid
Column: Waters BEH C4 10x1mm
Runtime: 18 minutes
Q-Exactive
LC/MS analysis

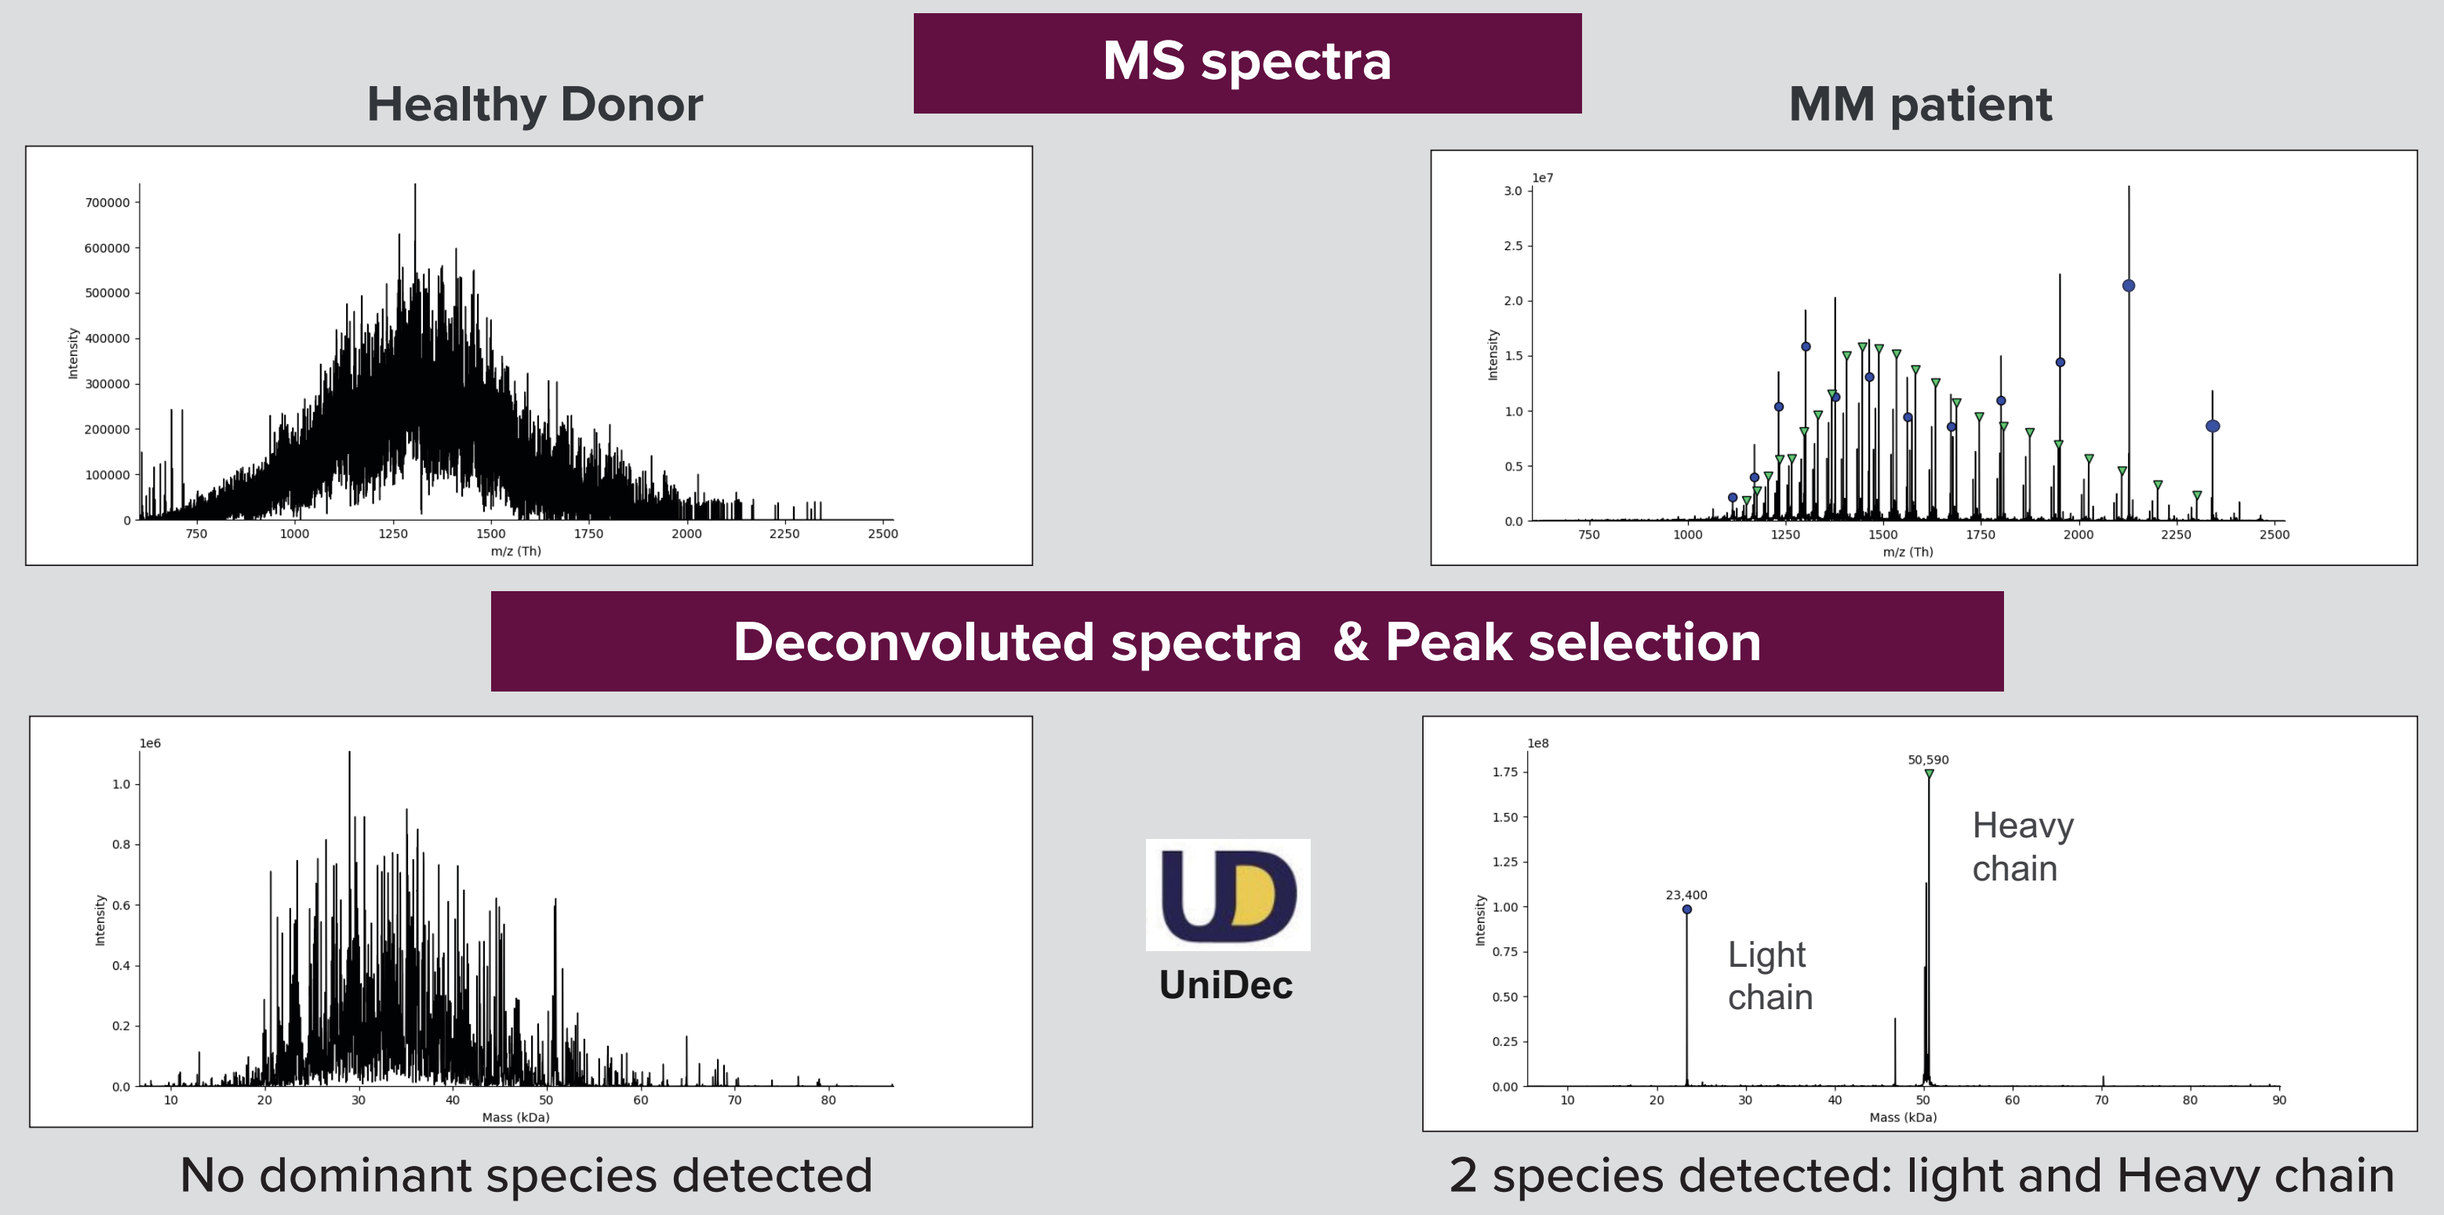

MS spectra
Healthy Donor
MM patient
Deconvoluted spectra & Peak selection
UniDec
Light chain
Heavy chain
No dominant species detected
2 species detected: light and Heavy chain

Figure 2. Analytical Workflow: deconvolution of MS spectra with UniDec[4] software for peak selection and plotting in samples from healthy donors (no dominant species) and MM patients (distinct signal generated by M-Protein).

ASSAY CHARACTERIZATION

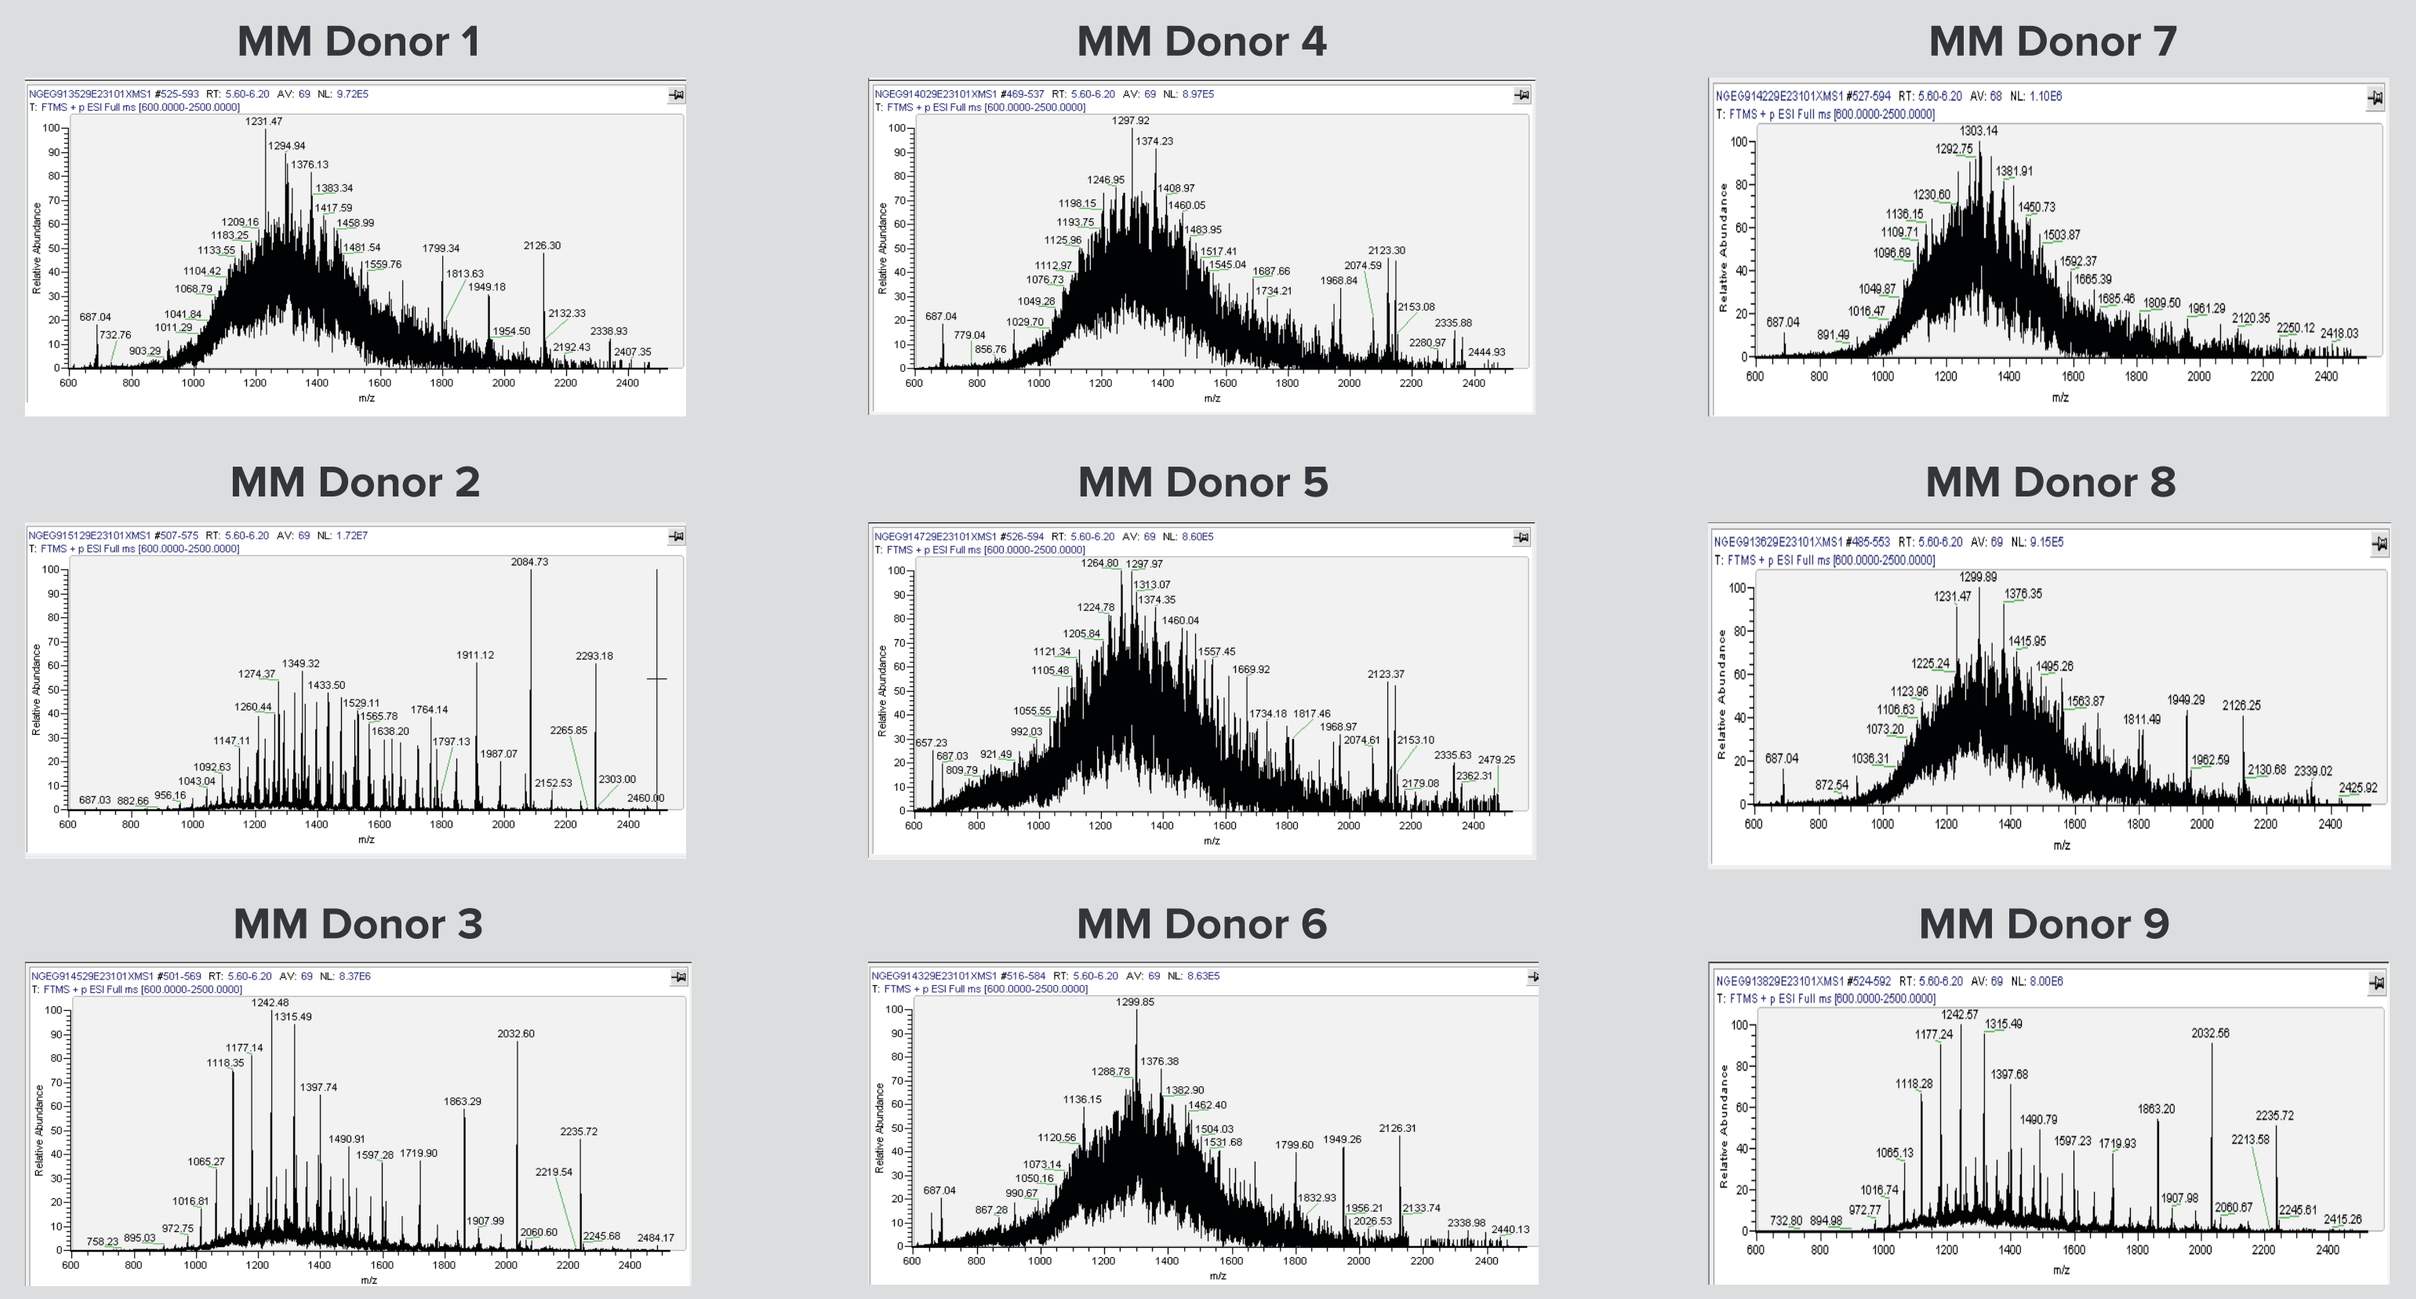

MM Donor 1
MM Donor 4
MM Donor 7
MM Donor 2
MM Donor 5
MM Donor 8
MM Donor 3
MM Donor 6
MM Donor 9

Figure 3. Nine examples of M-Protein responses in diagnosed multiple myeloma commercial samples.

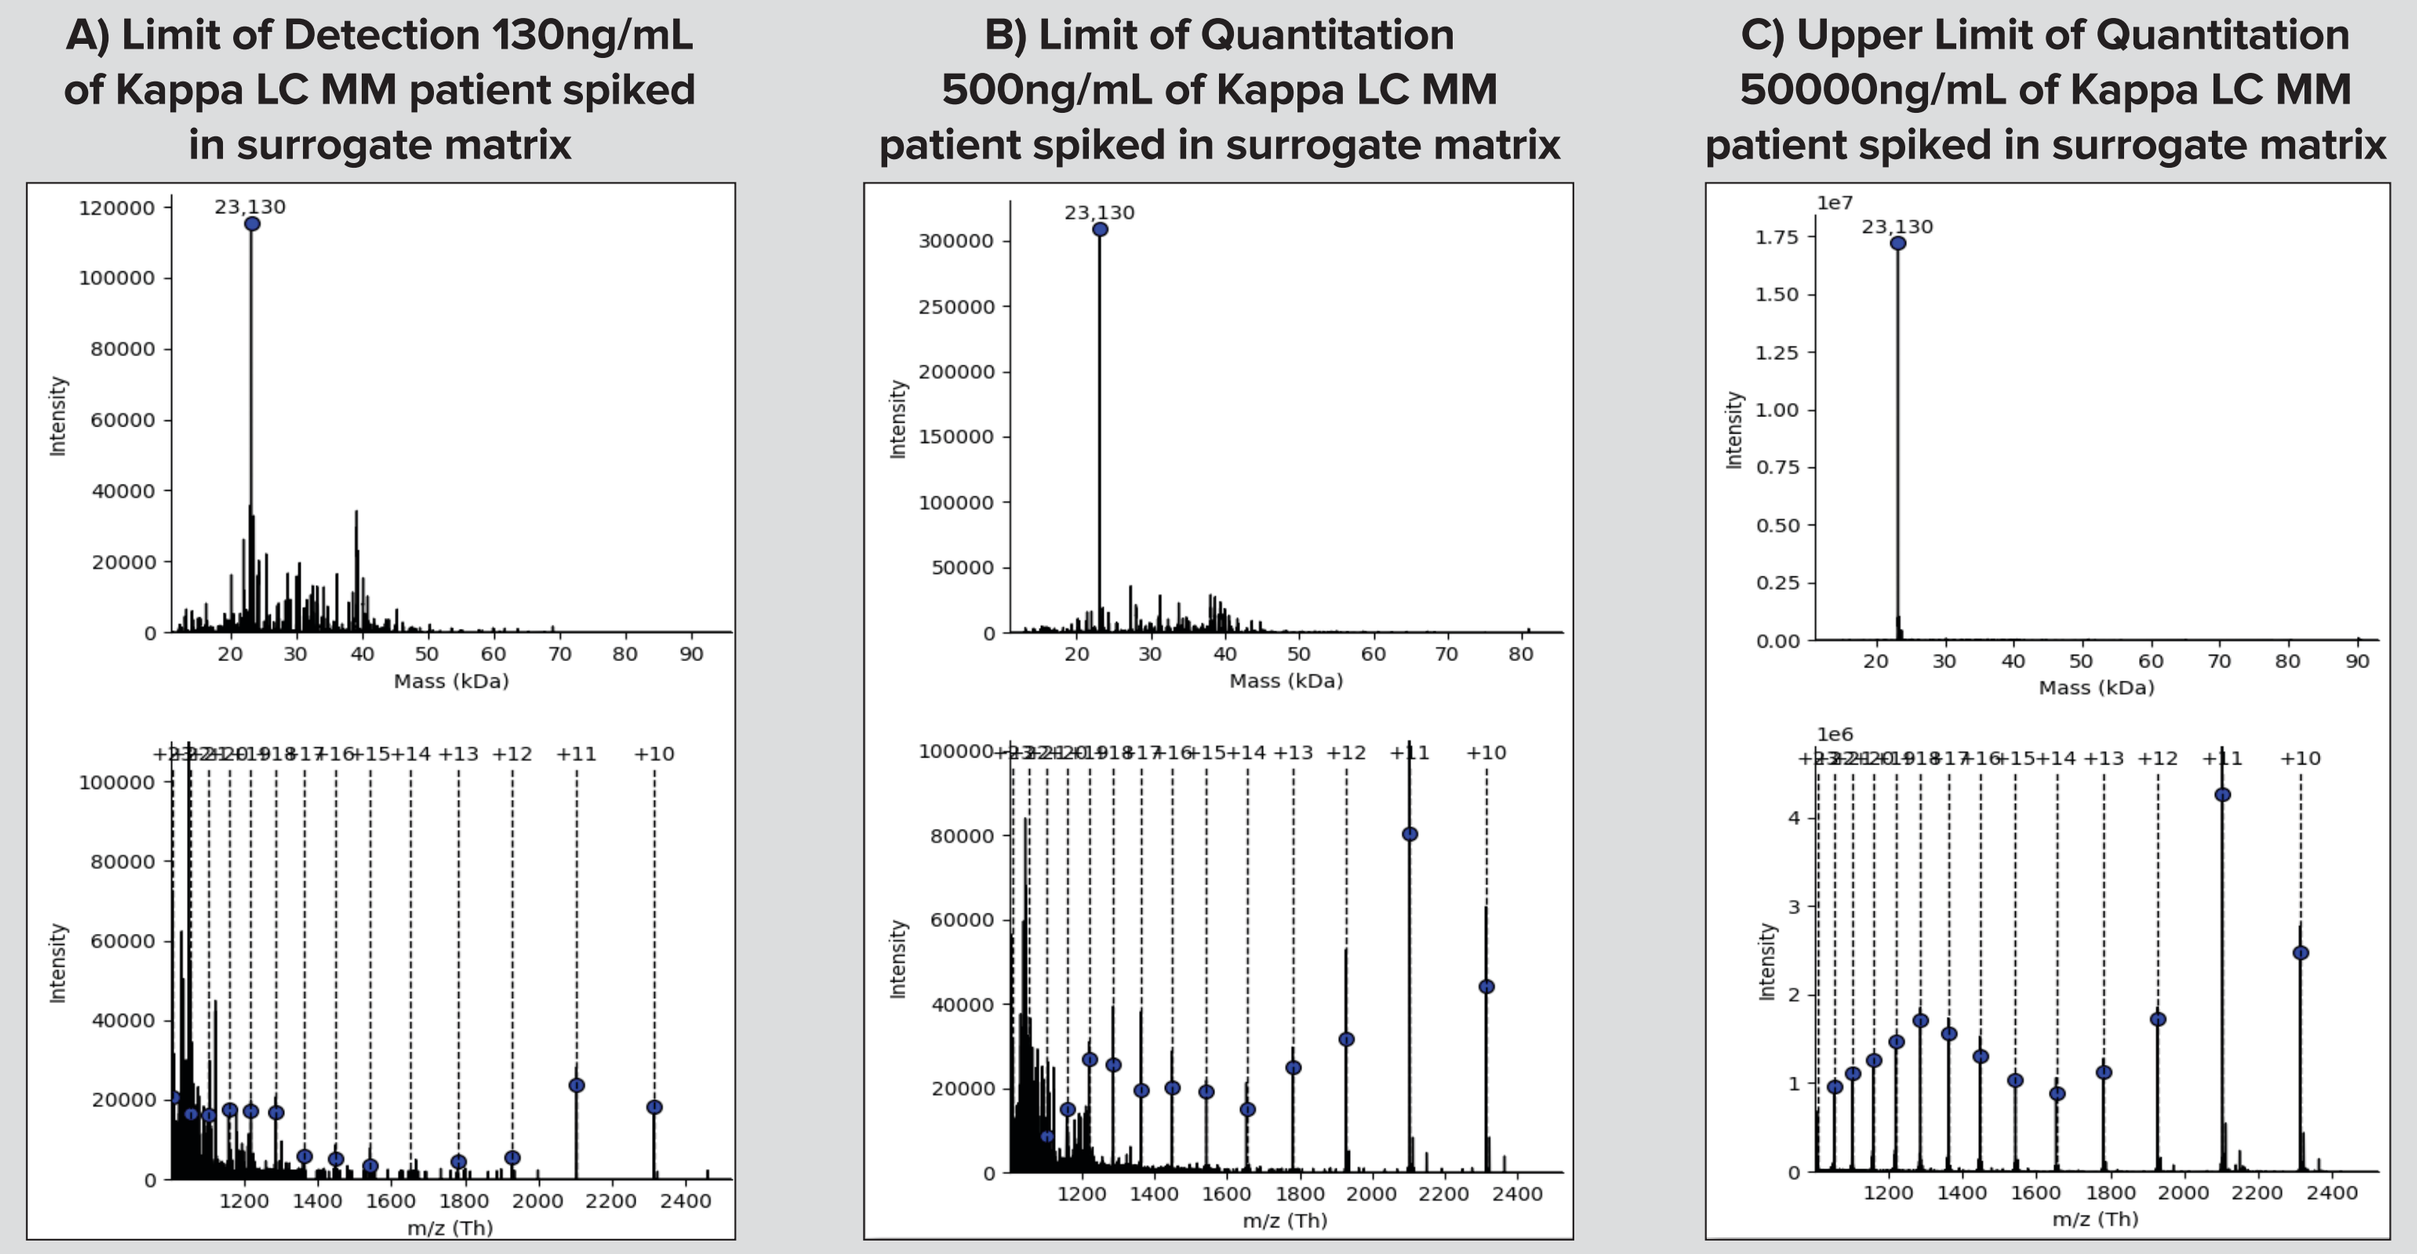

A) Limit of Detection 130ng/mL of Kappa LC MM patient spiked in surrogate matrix
B) Limit of Quantitation 500ng/mL of Kappa LC MM patient spiked in surrogate matrix
C) Upper Limit of Quantitation 50000ng/mL of Kappa LC MM patient spiked in surrogate matrix
23,130
Mass (kDa)
m/z (Th)
Intensity

Figure 4. Assessment of assay sensitivity. Zero-charge Mass Spectra (Top) and Data with Offset isolated species (bottom). **A)** Limit of Detection; **B)** Lower Limit of Quantitation and **C)** Upper limit of Quantitation.

PRECISION AND ACCURACY, ROBUSTNESS AND REPRODUCIBILITY

Table 1. Precision and accuracy run.

Melon Gel Sample Analysis	*Kappa Light Chain STD in Surrogate Matrix (µg/mL)*								*Kappa Light Chain QC in Surrogate Matrix (µg/mL)*			
	STD1	STD2	STD3	STD4	STD5	STD6	STD7	STD8	QC-LLOQ	QC-1	QC-2	QC-3
	0.50	**1.00**	**3.00**	**6.00**	**12.00**	**20.00**	**40.00**	**50.00**	**0.50**	**1.50**	**10.00**	**35.00**
Light chain	0.50	0.98	2.97	5.99	12.31	23.20	37.84	44.59	0.56	**1.93**	9.00	32.65
									0.53	1.75	10.31	35.73
									0.40	1.70	8.43	29.41
n	1	1	1	1	1	1	1	1	3	3	3	3
Mean	0.50	0.98	2.97	5.99	12.31	23.20	37.84	44.59	0.49	1.80	9.25	32.60
SD									0.08	0.12	0.96	3.16
% C.V.									17.1	6.6	10.4	9.7
% bias	0.9	-2.2	-1.0	-0.1	2.6	16.0	-5.4	-10.8	-1.1	19.8	-7.5	-6.9

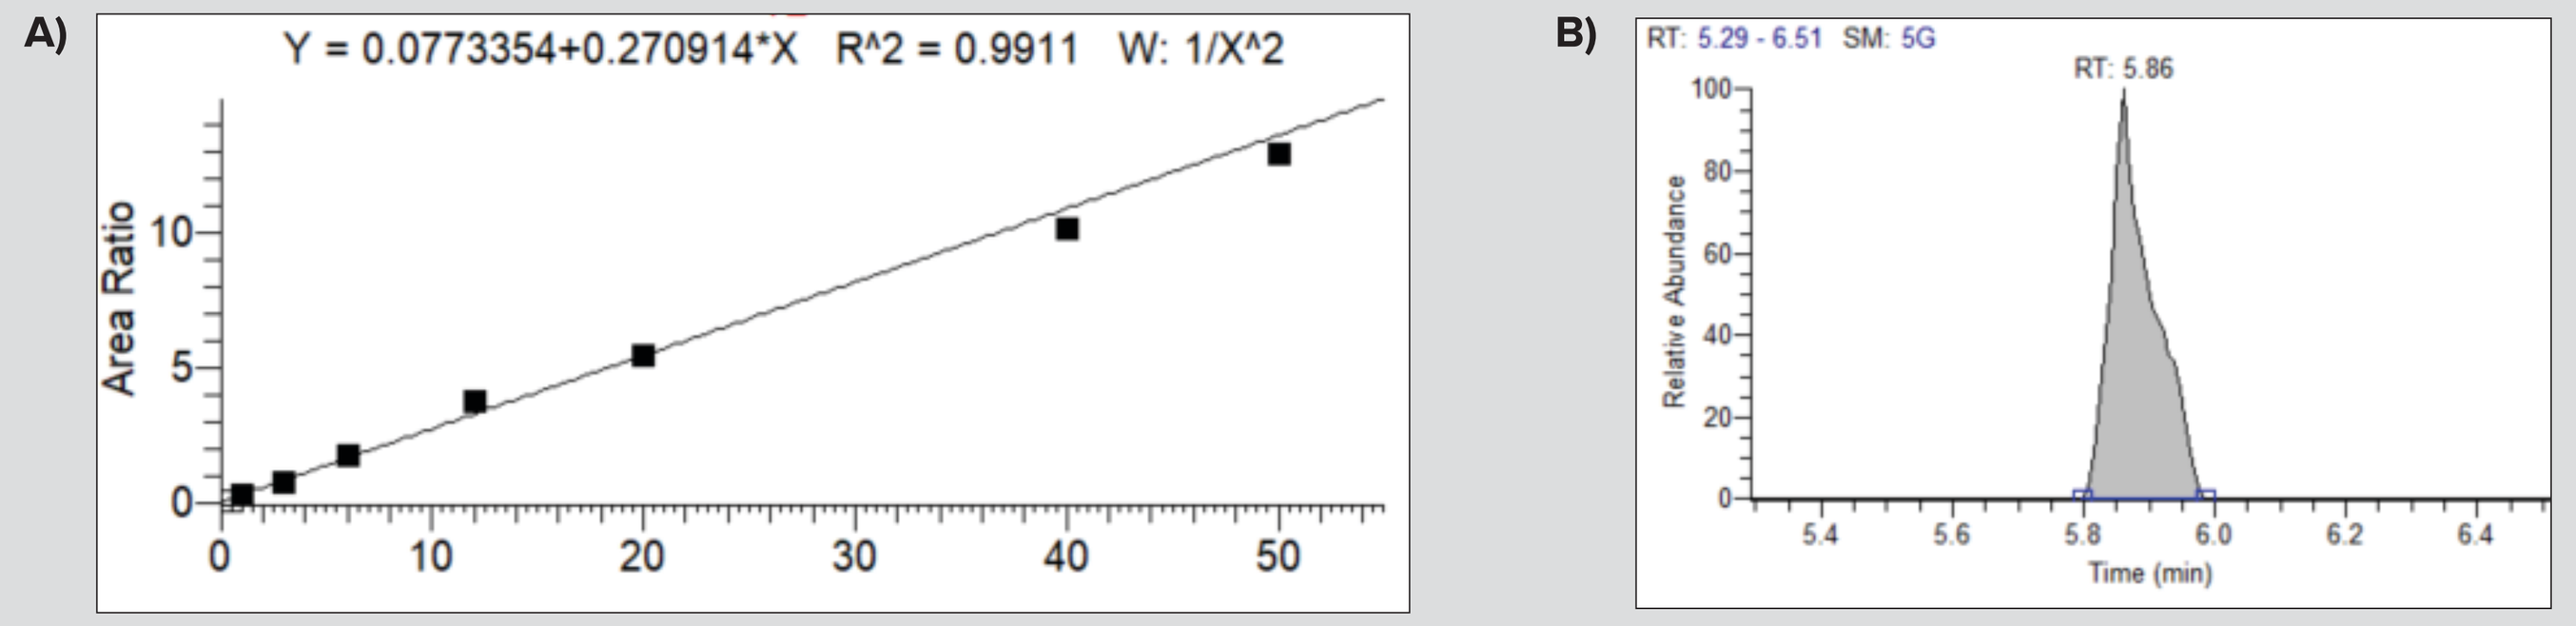

A)
Y = 0.0773354+0.270914*X R^2 = 0.9911 W: 1/X^2
Area Ratio
B)
RT: 5.29 - 6.51 SM: 5G
RT: 5.86
Relative Abundance
Time (min)

Figure 5. A) Standard curve generated by spiking Kappa sFLC spiked into surrogate matrix (PBS-BSA) at different concentrations; **B)** Extracted ion chromatogram for the lower limit of quantitation.

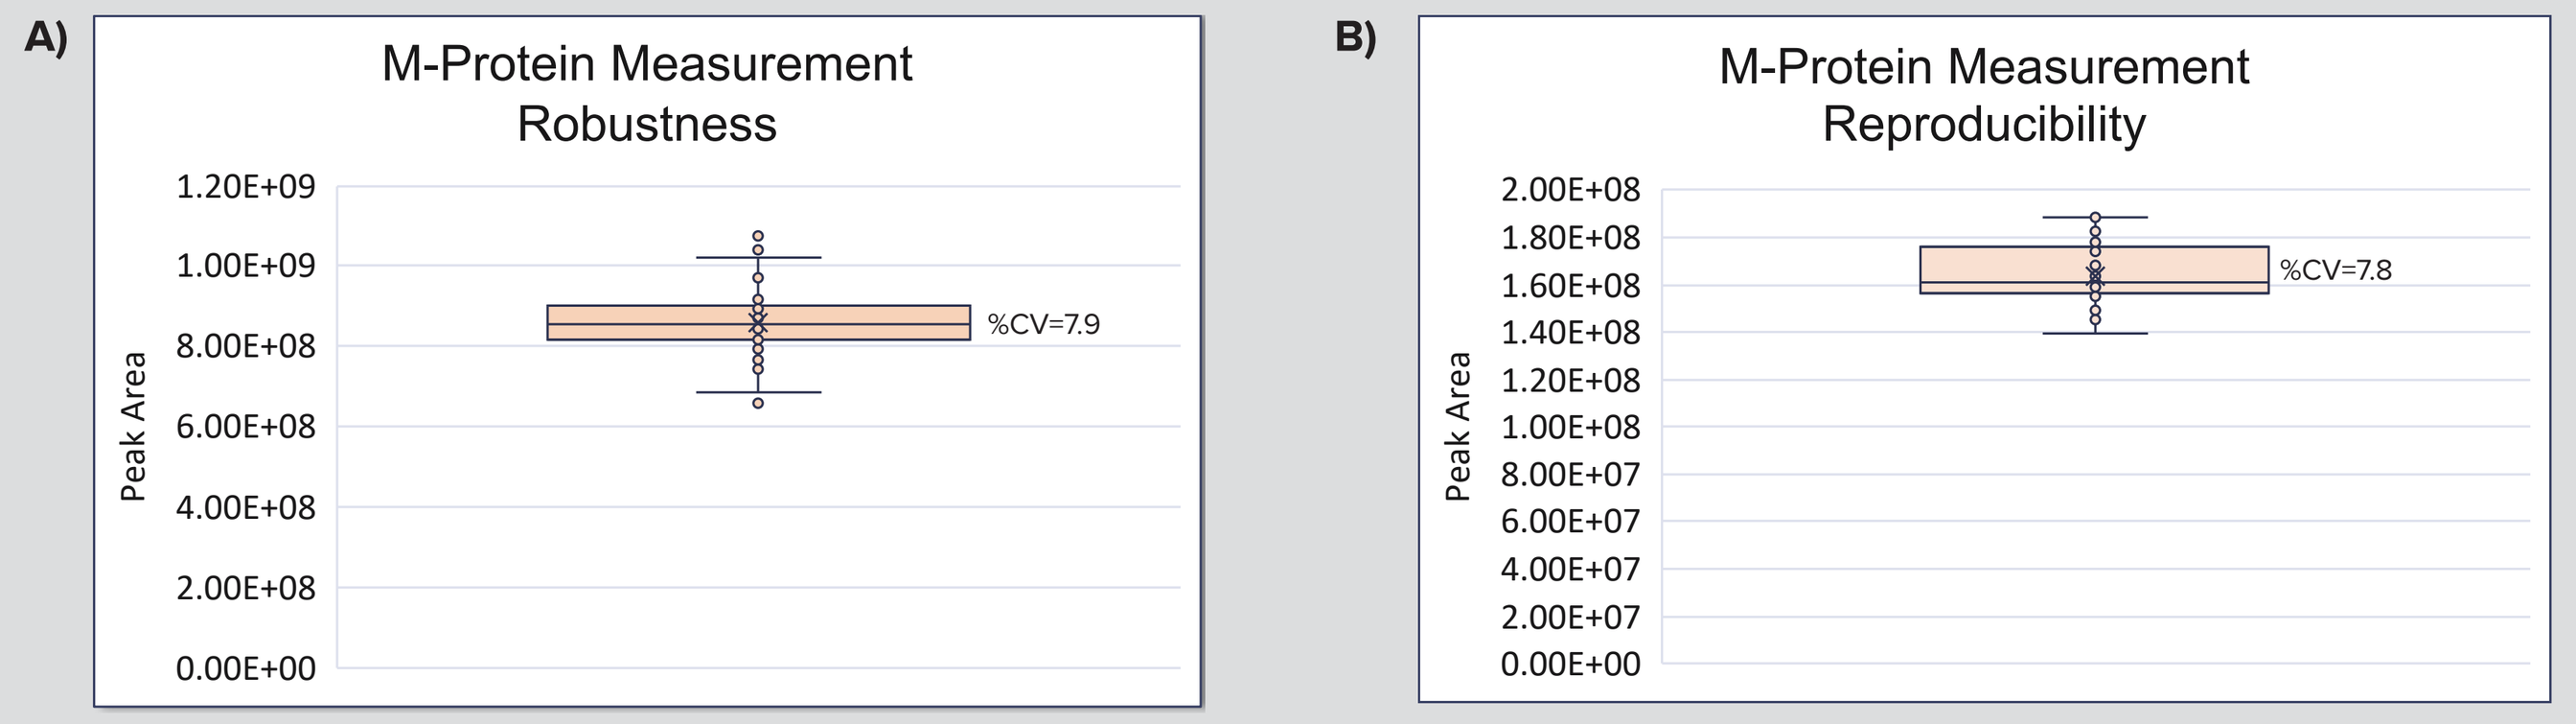

A)
M-Protein Measurement Robustness
Peak Area
%CV=7.9
B)
M-Protein Measurement Reproducibility
Peak Area
%CV=7.8

Figure 6. A) Method robustness tested on 96 injections of pooled samples extract after purification and reduction; **B)** Method reproducibility tested on 30 independent replicates with up to 5% hemolysis.

SENSITIVITY IN MATRIX

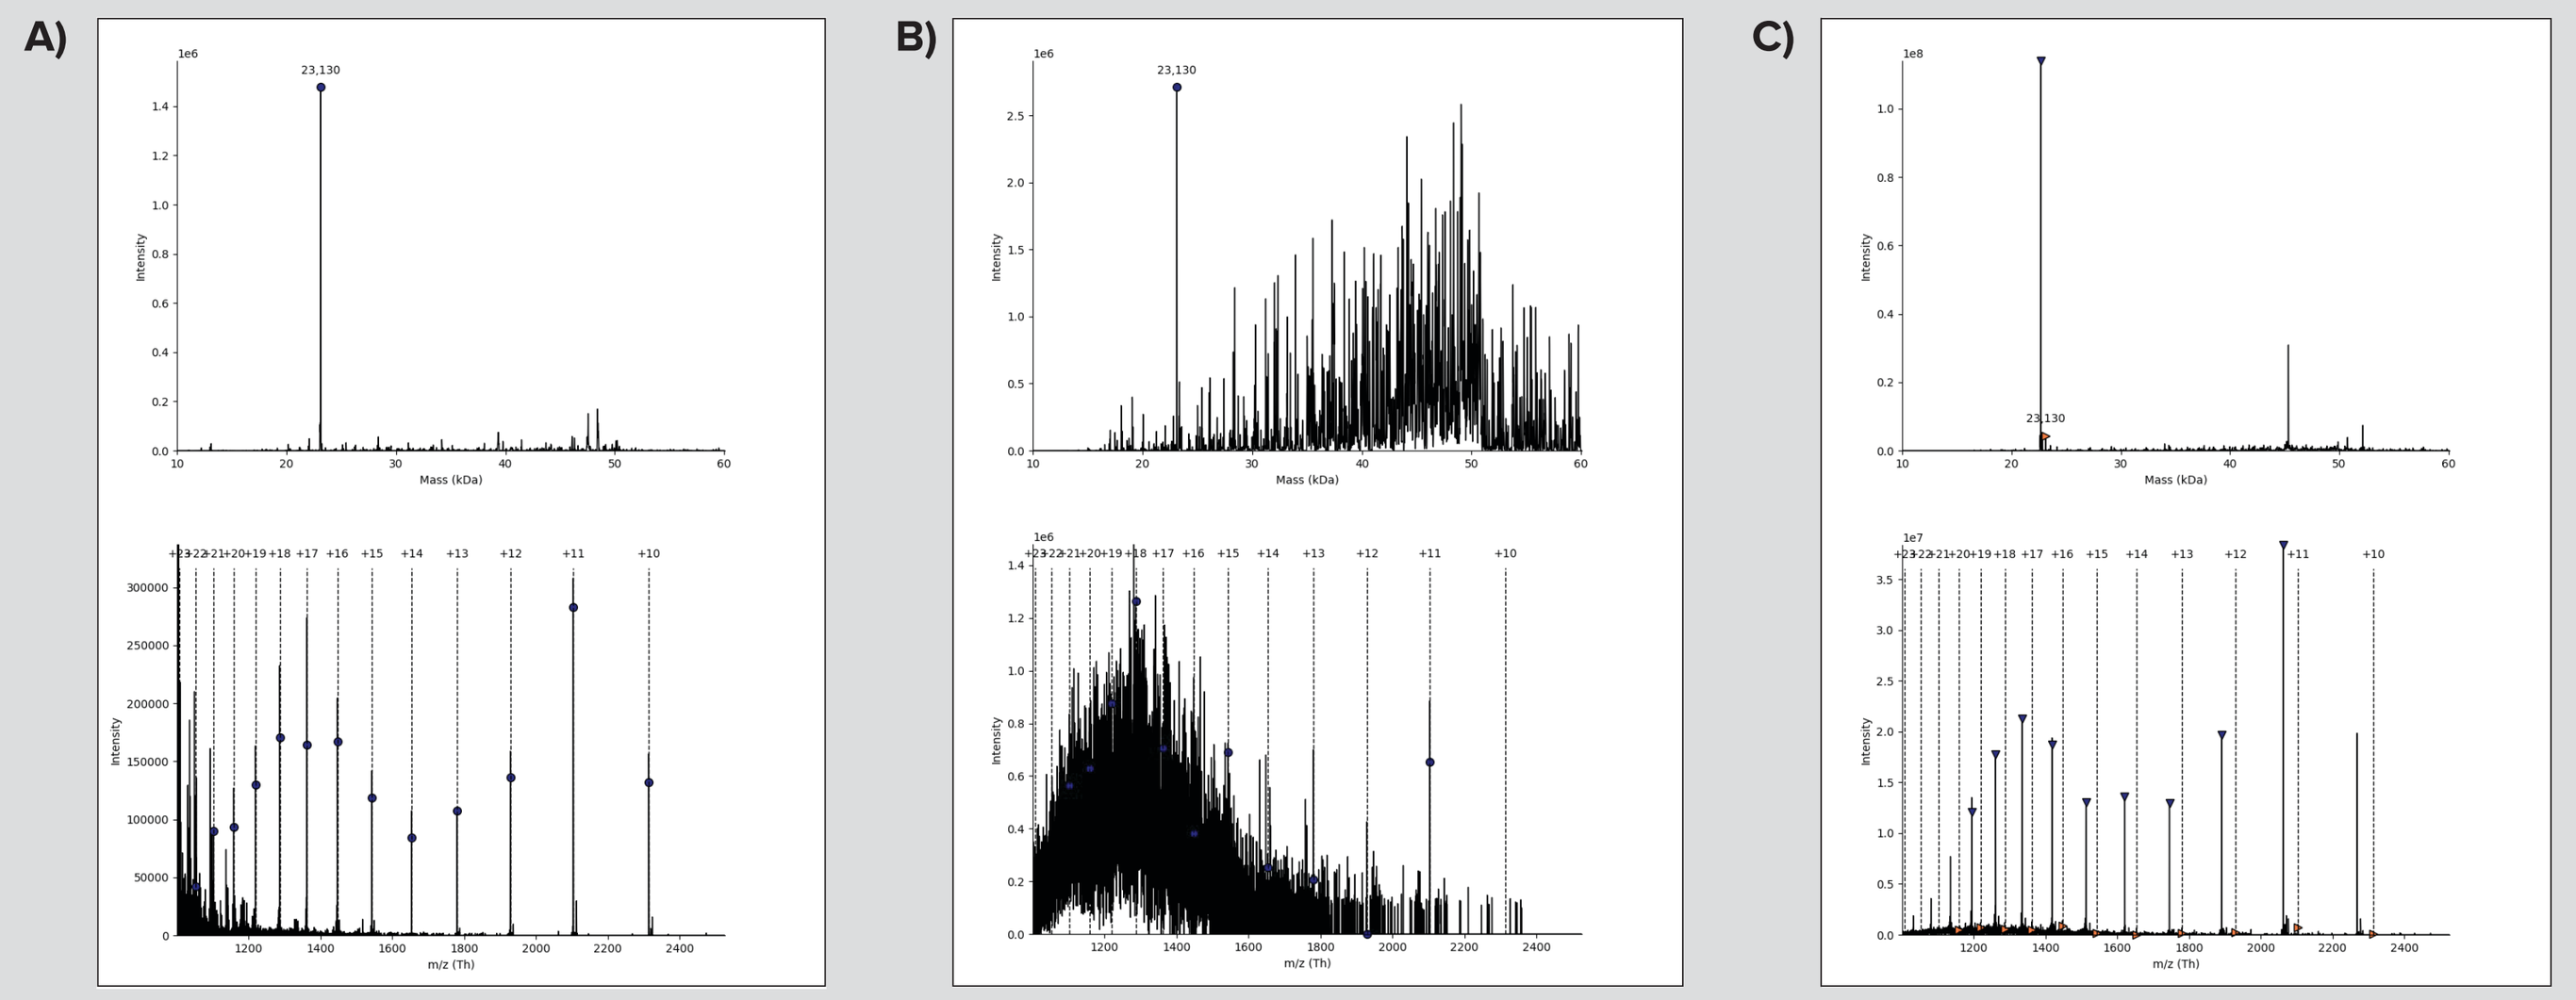

A)
B)
C)
23,130

Figure 7. Zero-charge Mass Spectra (Top) and Data with Offset isolated species (bottom). **A)** 5 µg of sFLC kappa standard spiked in surrogate matrix. **B)** 50 µg of sFLC kappa standard spiked in a healthy volunteer **C)** 50 µg of sFLC kappa standard spiked in a MM patient with high M-Protein level.

RESULTS IN CLINICAL SAMPLES

Table 2. Assessment of M-Protein intensity in plasma and serum over multiple timepoints.

Timepoint	Plasma Sample			Serum Sample			
	Light Chain Identified	Deconvoluted Mass	Peak Intensity	Light Chain Identified	Deconvoluted Mass	Peak Intensity	% Variation Plasma vs. Serum
T1	YES	23642	4.54E+07	YES	23642	4.84E+07	-6.2%
T2	YES	23642	4.18E+07	YES	23643	4.50E+07	-7.1%
T3	YES	23642	4.48E+07	YES	23643	4.42E+07	1.4%
T4	YES	23643	4.70E+07	YES	23642	4.39E+07	7.1%
T5	YES	23642	3.86E+07	YES	23643	4.38E+07	-11.9%
T6	YES	23643	4.38E+07	YES	23642	4.18E+07	4.8%
T7	YES	23642	4.78E+07	YES	23642	4.50E+07	6.2%

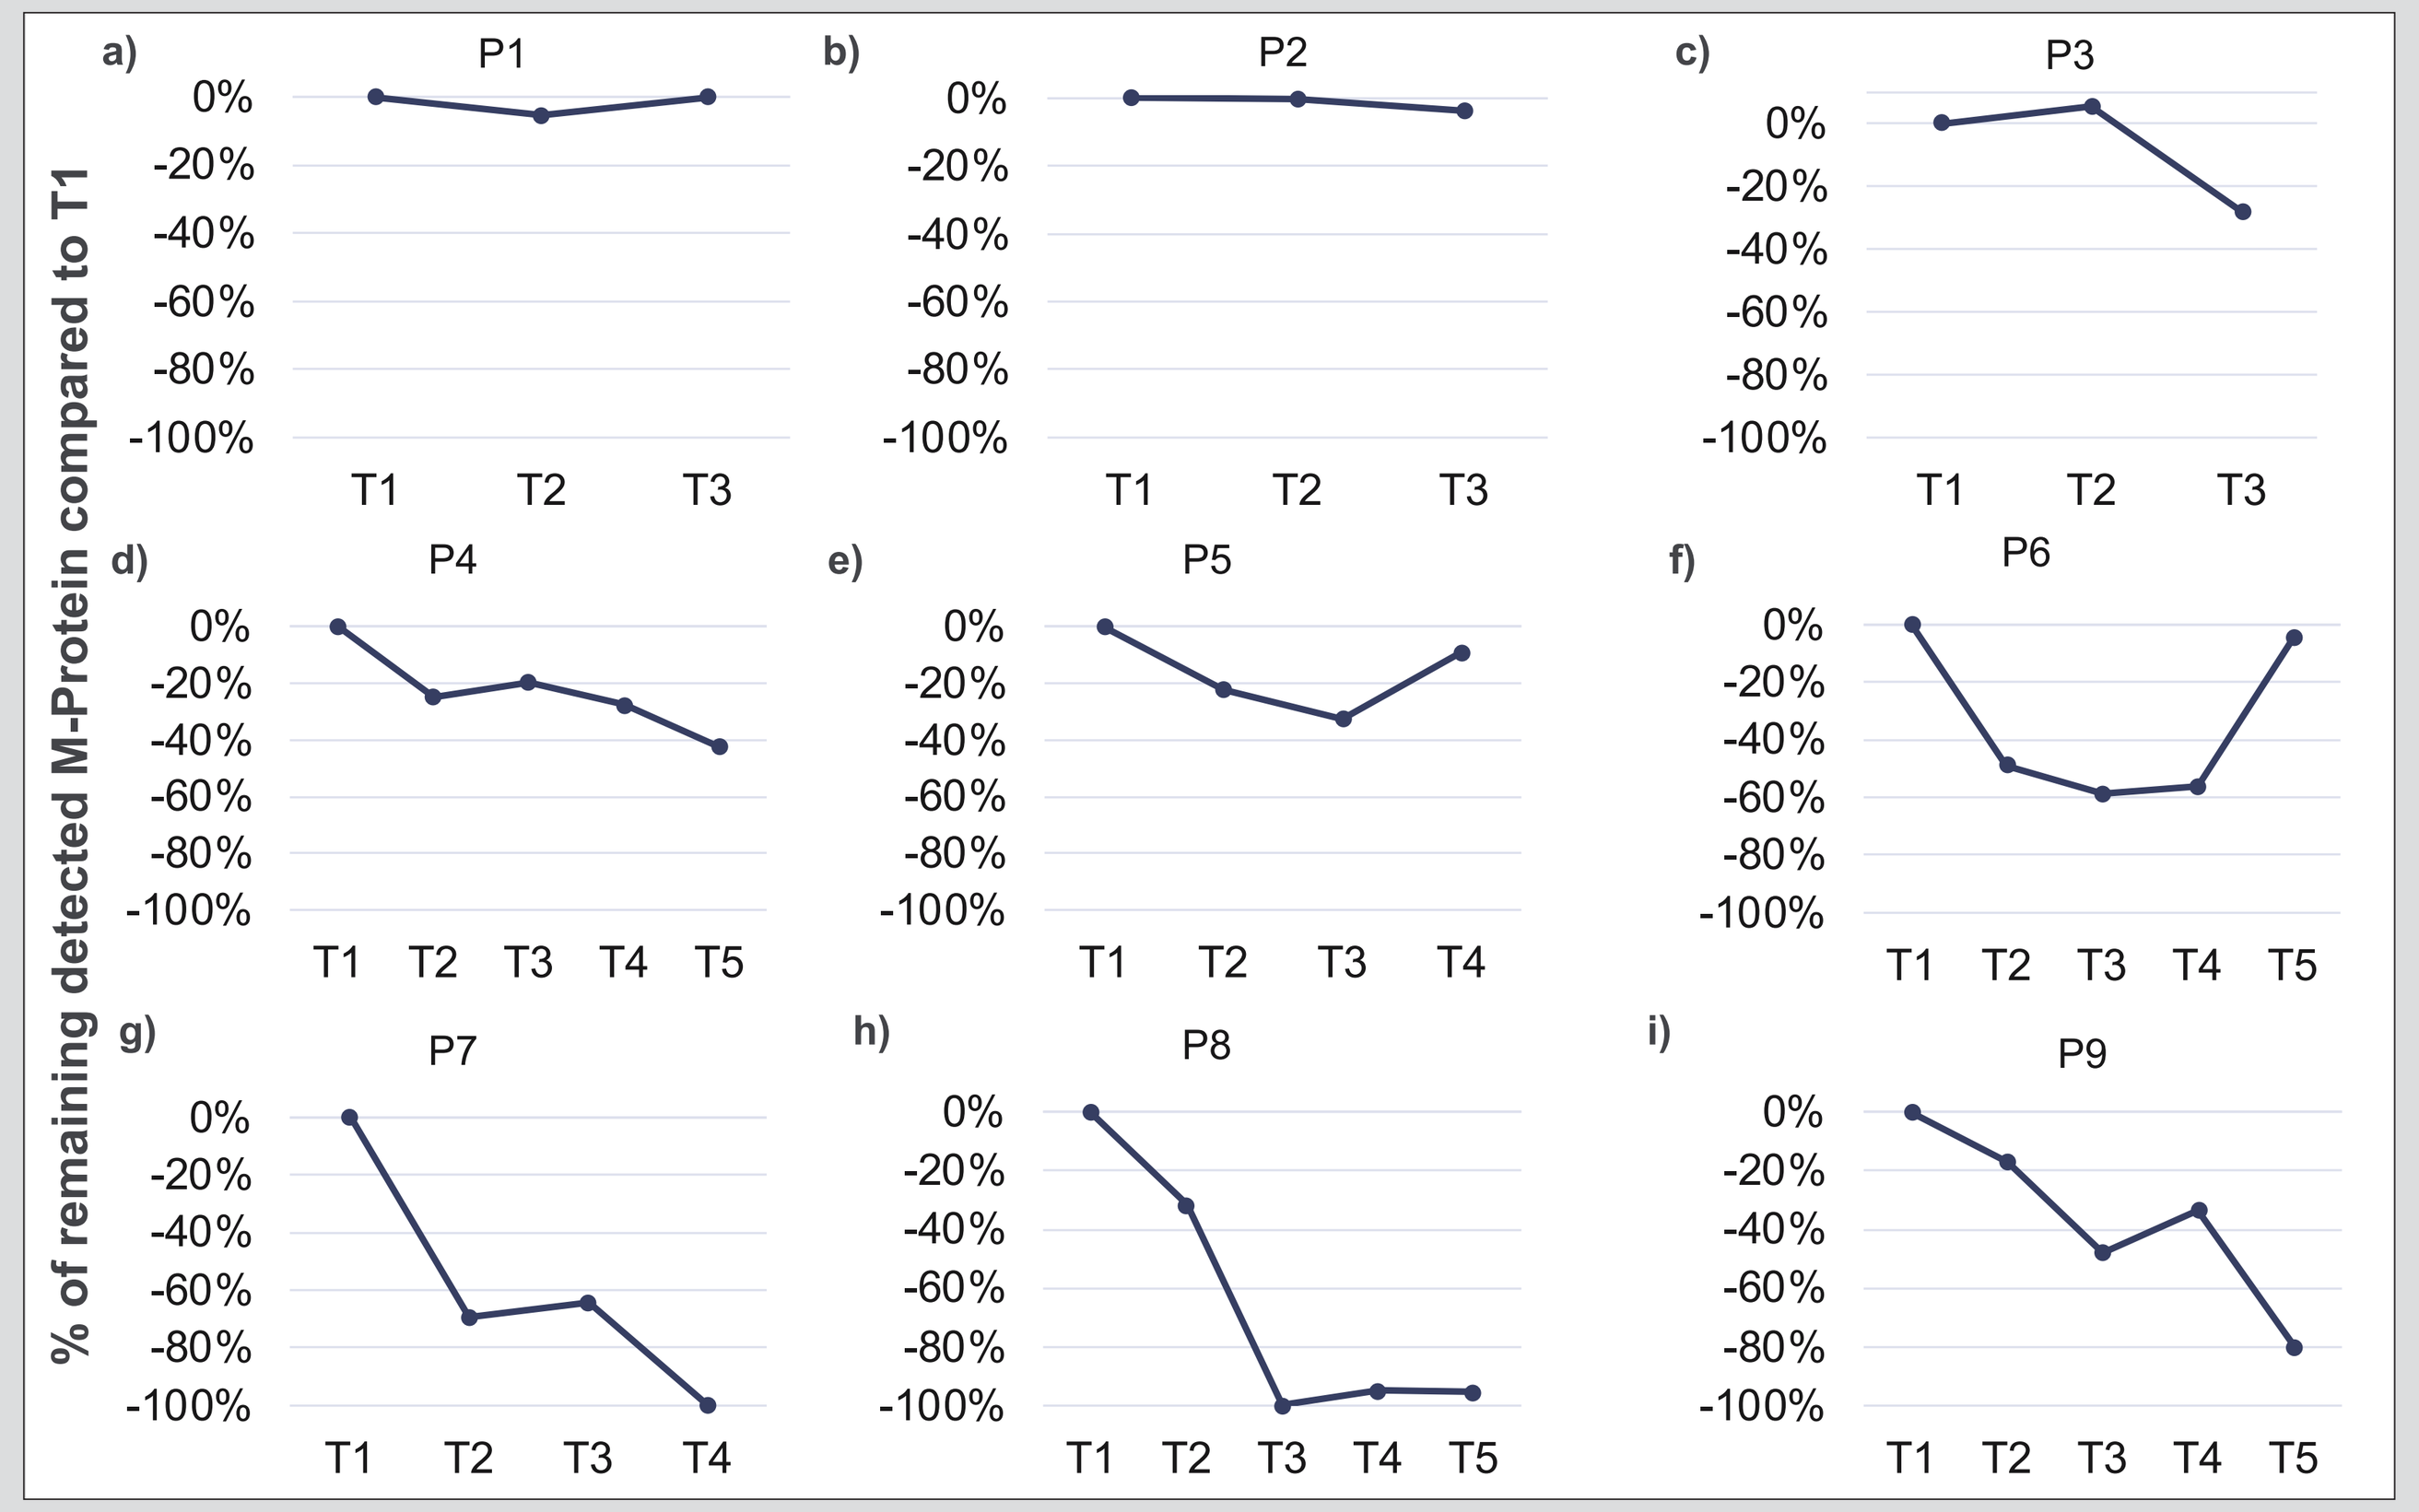

% of remaining detected M-Protein compared to T1
a) P1
b) P2
c) P3
d) P4
e) P5
f) P6
g) P7
h) P8
i) P9

Figure 8. M-Protein measurement over multiple timepoints (T1-T5) for nine different MM-patients (P1-P9).

CONCLUSION

- M-protein measurement can be achieved with a High-Resolution Mass Spectrometry workflow using just 10 µL of plasma/serum, reducing the need for painful BM-biopsy. This method is Sensitive, Precise & Accurate, Reproducible and Robust, despite the absence of an internal standard.
- Intact High-Resolution Mass Spectrometry currently offers the best sensitivity for measuring the IgG produced by the malignant cells with a fast assay process that can process up to 96 samples in approximately two hours.
- This method was successfully used to measure the M-protein levels of several multiple myeloma (MM) patients at different time points, showing a reduction in M-protein concentration by more than 90% in some cases. M-protein measurement by High Resolution Mass Spectrometry has the potential to enable high-frequency sampling of MM patients, providing a more comprehensive profile of disease progression.

REFERENCES

1 MONTEITH, Bethany E.; SANDHU, Irwindeep; LEE, Ann S. Management of multiple myeloma: a review for general practitioners in oncology. Current Oncology, 2023, 30.5: 4382-4401.
2 PERROT, Aurore, et al. Minimal residual disease negativity using deep sequencing is a major prognostic factor in multiple myeloma. Blood, The Journal of the American Society of Hematology, 2018, 132.23: 2456-2464.
3 DERMAN, Benjamin A., STEFKA, Andrew T., JIANG, Ken, et al. Measurable residual disease assessed by mass spectrometry in peripheral blood in multiple myeloma in a phase II trial of carfilzomib, lenalidomide, dexamethasone and autologous stem cell transplantation. Blood Cancer Journal, 2021, vol. 11, no 2, p. 19.
4 MARTY, Michael T., et al. Bayesian deconvolution of mass and ion mobility spectra: from binary interactions to polydisperse ensembles. Analytical chemistry, 2015, 87.8: 4370-4376.

Please send inquiries to
Luca Genovesi, Ph.D.
R&D Group Leader
luca.genovesi@Cellcarta.com

Day3-P24

www.cellcarta.com