4051

SUCCESSFUL MONITORING OF CHECKPOINT INHIBITORS BY FLOW CYTOMETRY

Scott Sugden, Laïla-Aïcha Hanafi, Valérie Hébert, Delphine Labit, Vicky Sgouroudis, Jean-François Poulin and Philippe Pouliot

Caprion Biosciences Inc.

CAPRION

INTRODUCTION

Cancer immunotherapies represent more than half of treatments approved for oncology, as well as those currently in development. Among the key targets of these therapies are immune-checkpoint molecules expressed on T cells (e.g. PD-1, LAG-3) or their ligands (e.g. PD-L1). Novel immunotherapies targeting these molecules are designed to circumvent immune-suppression and reactivate anti-tumor response.

Flow cytometry is an information-rich method for cellular analysis. In the age of immunotherapy, the importance of flow cytometry in drug development has increased dramatically. Flow cytometry panels can be used at different stage of a drug development from R&D to late clinical studies. The characterization and monitoring of circulating and tumor infiltrating immune cells by flow cytometry is a critical aspect of pre-clinical and clinical drug development. In order to generated robust data to support drug development, flow cytometric methods must be fully optimized based on their intended purpose of use.

In this work, we have screened antibodies for PD-1 expression on CD4+ and CD8+ T cells. PD-1 is expressed at low levels on the surface of resting T lymphocytes and is upregulated via signaling through the T-cell receptor (TCR). For this we tested the expression on freshly-thawed PBMC and SEB-activated PBMC.

Secondly, we have screened antibodies for LAG-3 and PD-L1 expression in whole blood collected in Cyto-Chex. Both immune-regulatory molecules are expressed at only low levels in healthy donor blood. For this reason, LAG-3 antibodies were screened on whole blood spiked with SEB-activated for 4 days which leads to the upregulation of LAG-3 due to exhaustion of the cells. For PD-L1, internal data have demonstrated that when cells are rested for at least 4 days at 37°C, the expression of PD-L1 is induced on monocytes and T cells. Rested PBMC have been spiked in whole blood for the screening of anti-PD-L1 antibodies.

PURPOSE

The purpose of the work presented in this poster was to identify appropriate flow cytometry antibody clones to detect PD-1, LAG-3 and PD-L1 for development of immune-monitoring panels.

OBJECTIVES

- Screen different clones for the detection of PD-1, LAG-3 and PD-L1.
- Better characterize the basal and induced PD-1 expression on CD4+ and CD8+ T cells

METHODS

1) Staining Methodology

Cryopreserved PBMC

For PD-1 staining, the testing was performed for staining on PBMC. Cells were stained at 1x106 cells per well first with FcR block for 10 minutes at RT. After wash, cells were stained for 30 minutes at 4°C with the antibody cocktail. Finally cells were fixed for 30 minutes with PFA at 4°C before acquisition on the cytometer.

Whole Blood

For LAG-3 and PD-L1 staining, the testing was performed for staining on whole blood collected in Cyto-chex. FcR block was added to whole blood collected from healthy donor and incubated for 10 minutes at RT. Antibodies were added and stained for 30 minutes at 4°C. Finally, lyse/fix solution was added, and blood was washed prior to acquisition on the cytometer.

2) Clones and Conditions Tested

PD-1

Different PD-1 clones were investigated on healthy donor cryopreserved PBMC using a surface staining method. To assess anti-PD-1 clones, unstimulated PBMC and SEB-activated PBMC were used to compare 5 different clones from various manufacturers. PD-1 staining was assessed in CD4+ and CD8+ T cells.

Table 1: Anti-PD-1 clone tested

Clone	Manufacturer	Catalog #
EH12.2H7	Biolegend	329906
EH12.1	BD Biosciences	560795
eBioJ105	eBioscience	12-2799-42
MIH4	BD Biosciences	5557946
MIH4	eBioscience	12-9969-42

LAG-3

LAG-3 detection was investigated on whole blood collected in Cyto-Chex tubes. To assess anti-LAG-3 clones, whole blood collected from healthy donor were spiked or not with SEB-activated PBMC to induce expression of LAG-3.

Table 2: Anti-LAG-3 clone tested

Clone	Manufacturer	Catalog #
3DS223H	eBioscience	12-2239-42
7H2C65	Biolegend	369206
11C3C65	Biolegend	369306
REA351	Miltenyi	130-105-452
T47-530	BD Biosciences	565616

PD-L1

PD-L1 detection was investigated on whole blood collected in CytoChex tubes. To assess anti-PD-L1 clones, whole blood collected from healthy donor were spiked or not with PBMC rested for 4 days to induce expression of PD-L1.

Table 3: Anti-PD-L1 clone tested

Clone	Manufacturer	Catalog #
130021	R&D System	FAB1561A-100
E1L3N	Cell Signaling	15005
MIH1	BD Biosciences	563741
PD-L1	Beckman Coulter	B36126

RESULTS

Figure 1: Gating strategy for identification of CD4+ and CD8+ T cells for PD-1 assessment.

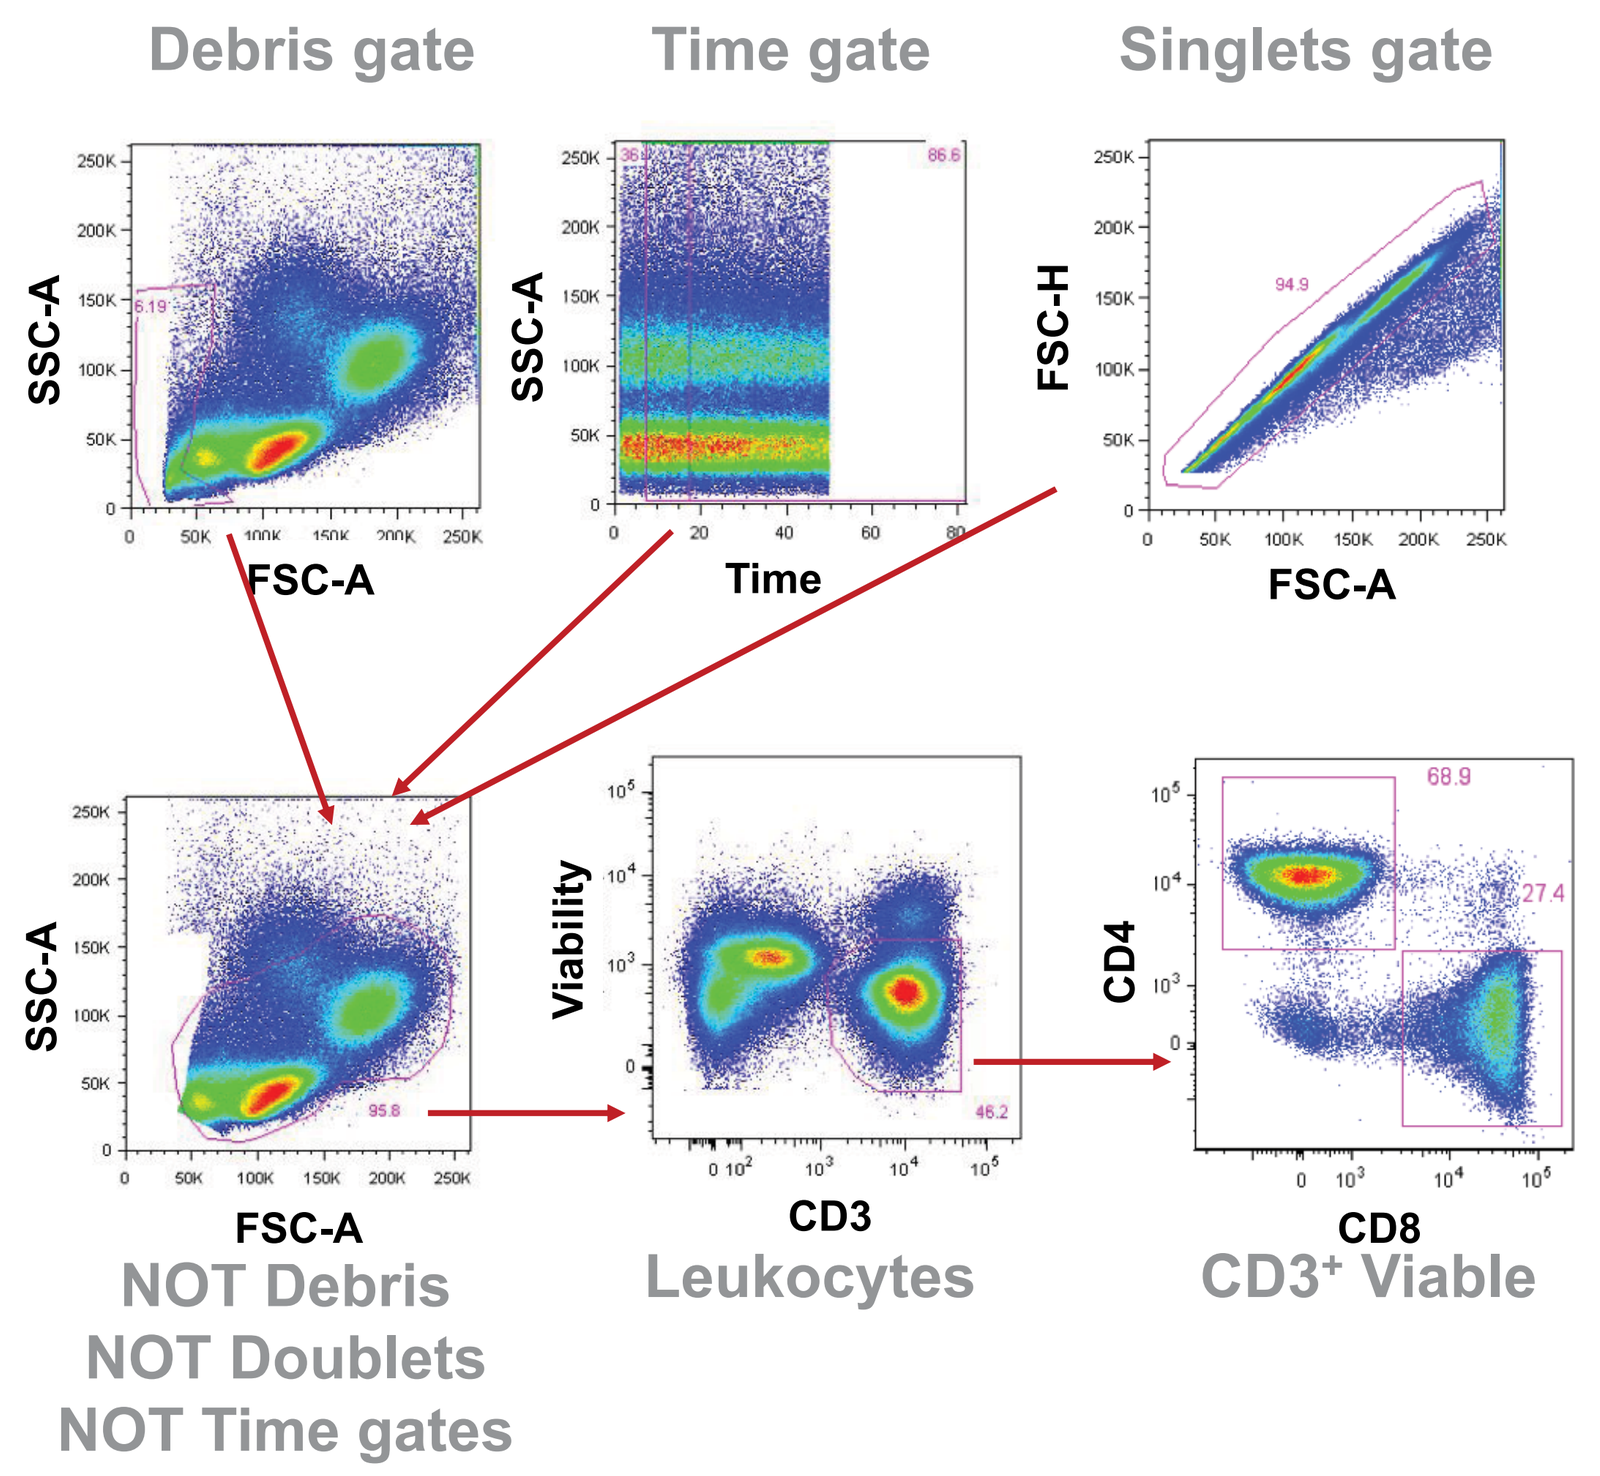
Debris gate
Time gate
Singlets gate
SSC-A
FSC-A
Time
FSC-H
Viability
CD3
CD4
CD8
NOT Debris
NOT Doublets
NOT Time gates
Leukocytes
CD3+ Viable

Figure 2: Anti-PD-1 clone screening on stimulated and non-stimulated PBMC.

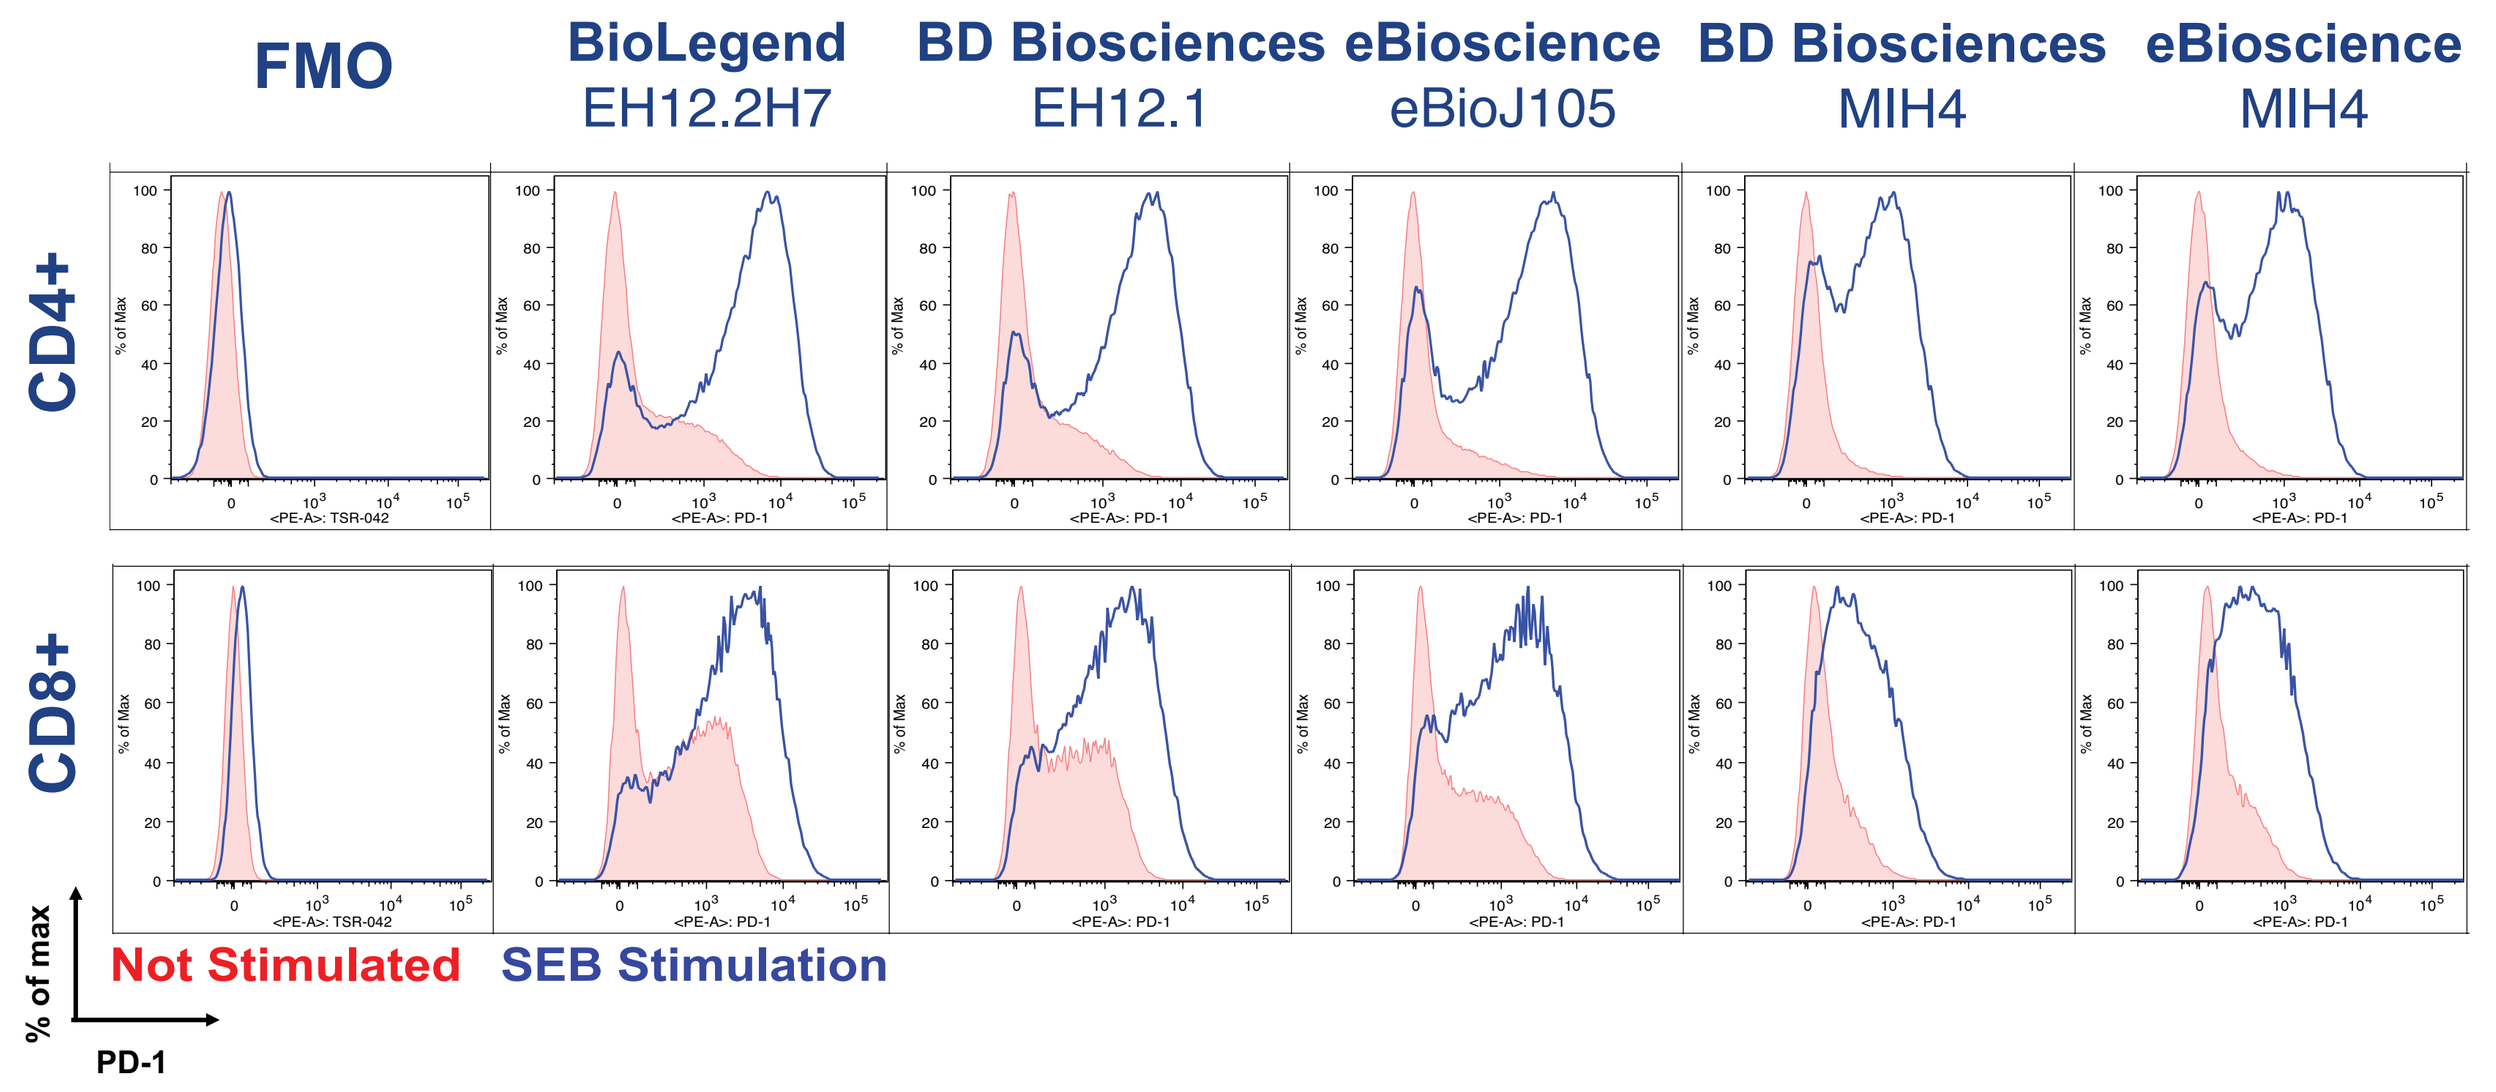
FMO
BioLegend EH12.2H7
BD Biosciences EH12.1
eBioscience eBioJ105
BD Biosciences MIH4
eBioscience MIH4
CD4+
CD8+
% of max
PD-1
Not Stimulated
SEB Stimulation

Figure 3: Detection of PD-1 expression on freshly thawed or SEB-activated PBMC varies widely depending on detection antibody clone.

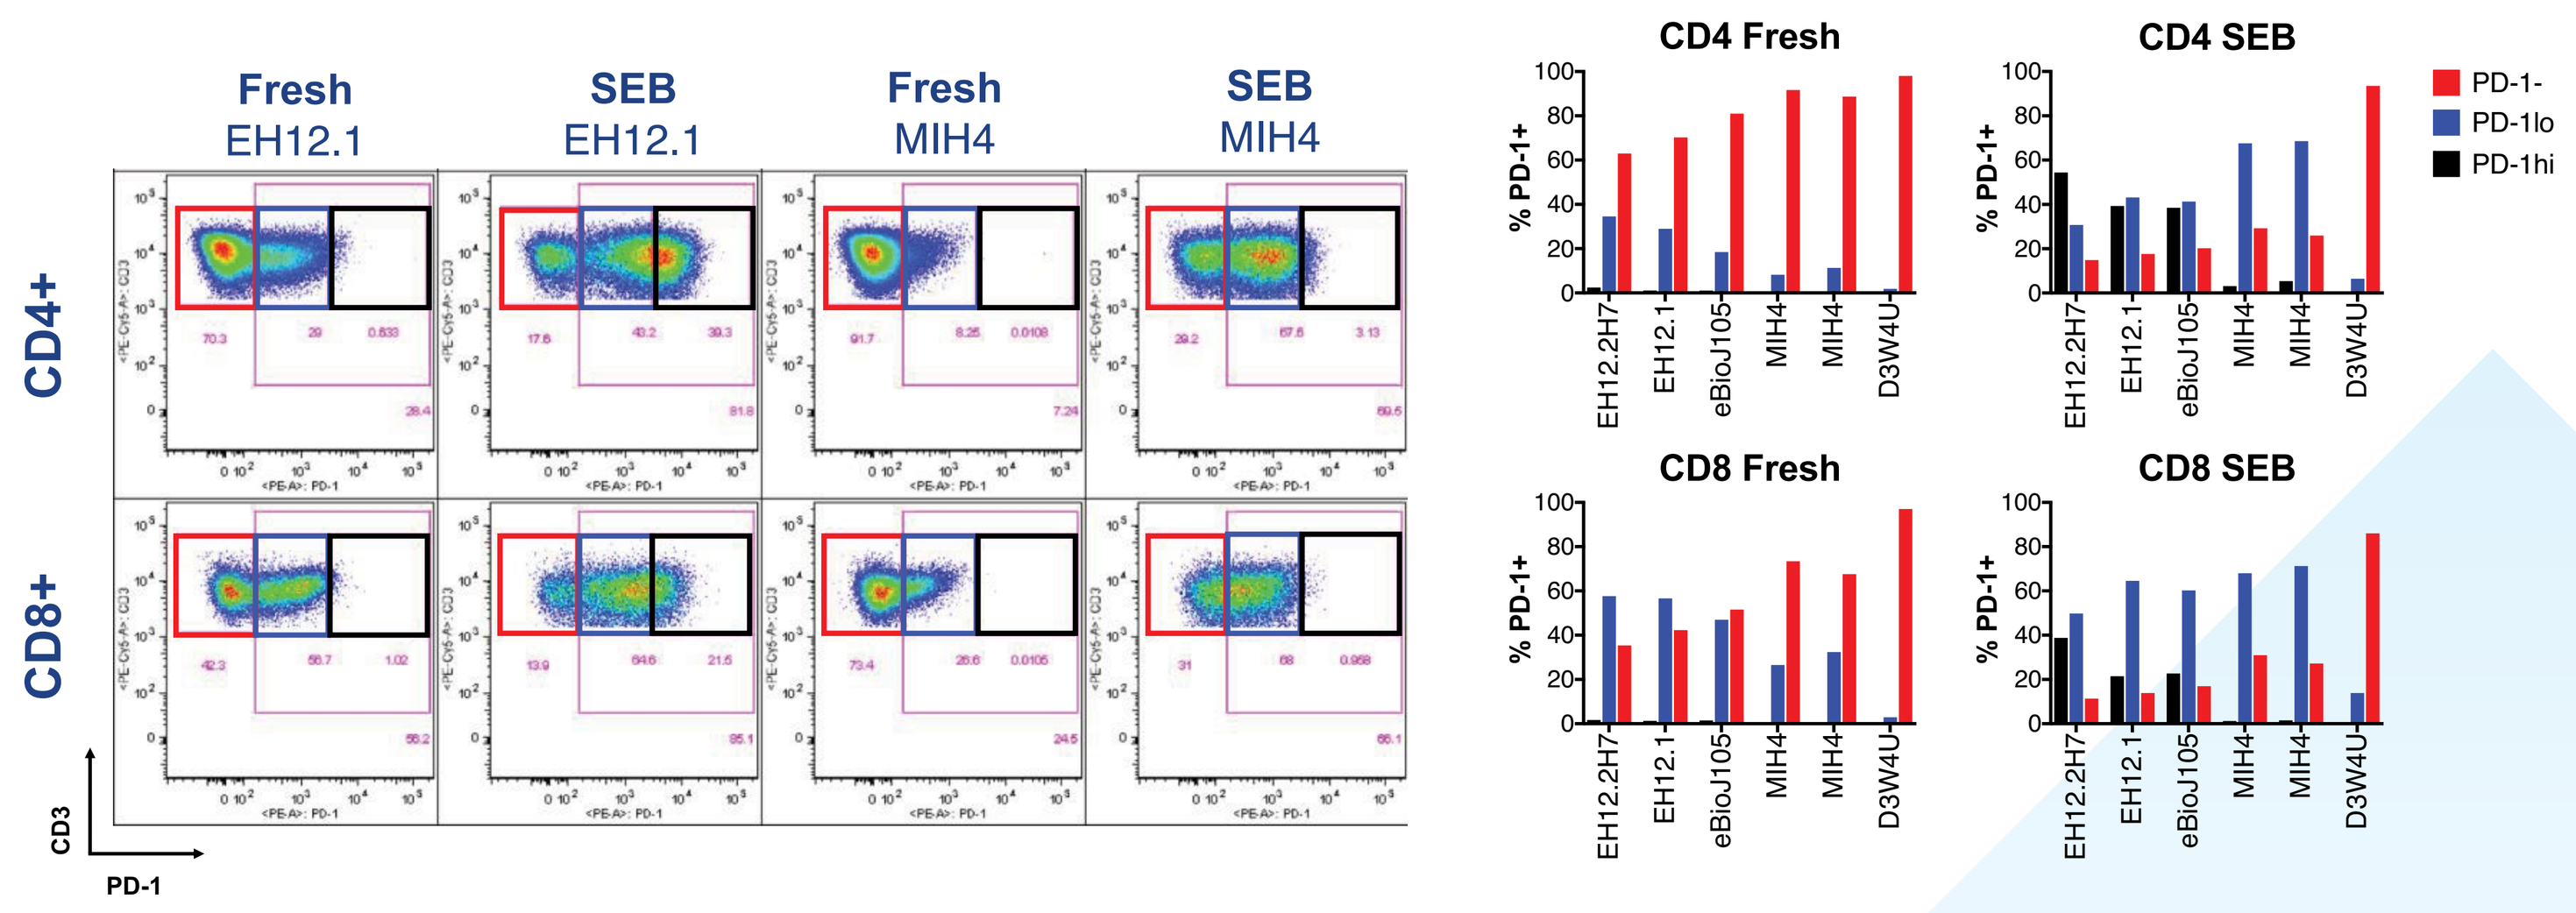
Fresh EH12.1
SEB EH12.1
Fresh MIH4
SEB MIH4
CD4+
CD8+
CD3
PD-1
CD4 Fresh
CD4 SEB
CD8 Fresh
CD8 SEB
% PD-1+
EH12.2H7
EH12.1
eBioJ105
MIH4
MIH4
D3W4U
PD-1-
PD-1lo
PD-1hi

Figure 4: Anti-LAG-3 clone screening on Cyto-Chex spiked or not with activated PBMC.

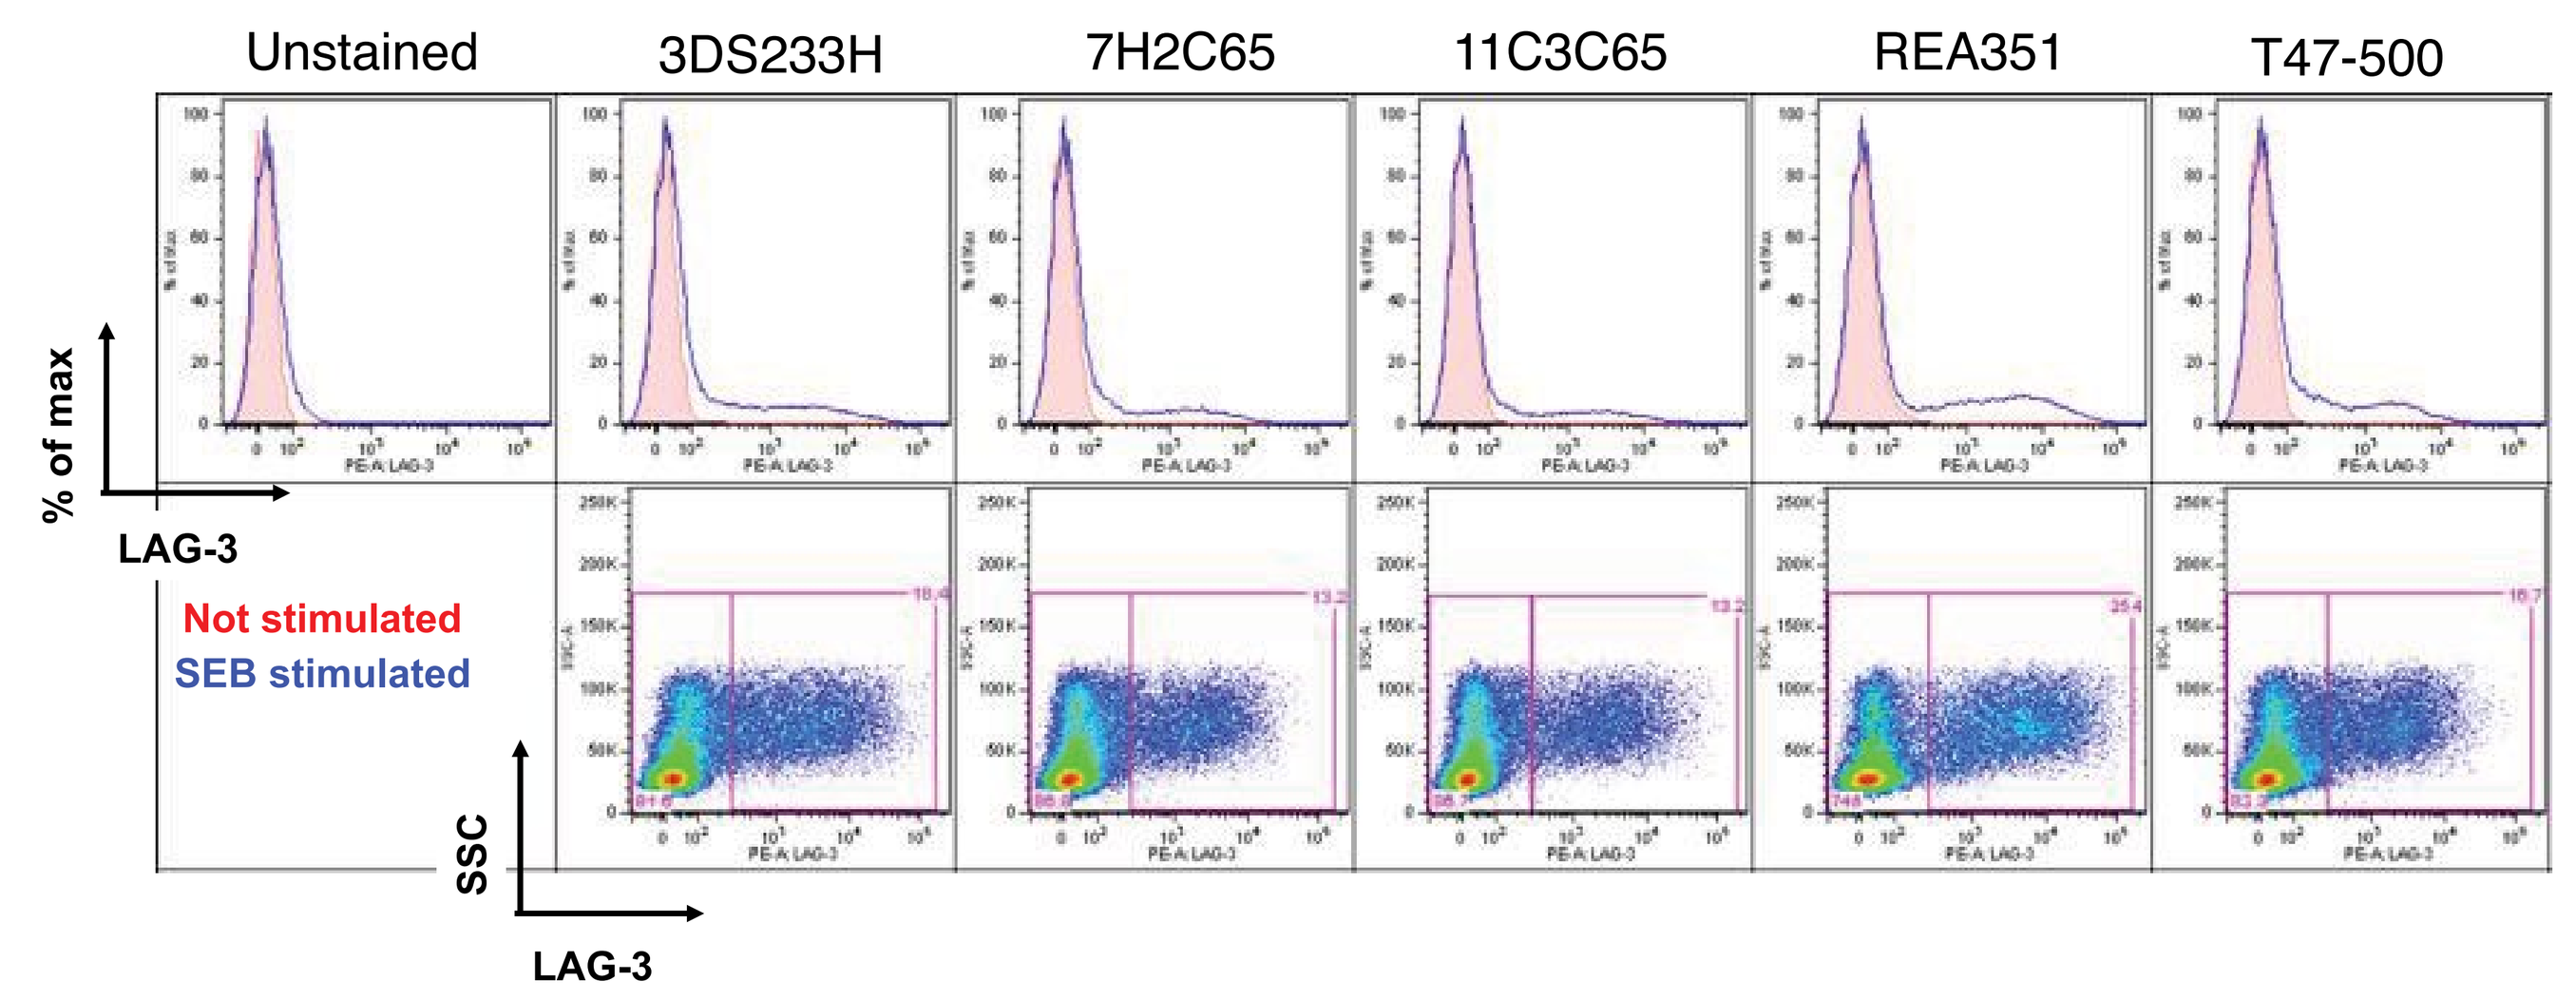
Unstained
3DS233H
7H2C65
11C3C65
REA351
T47-500
% of max
LAG-3
Not stimulated
SEB stimulated
SSC
LAG-3

Figure 5: Anti-PD-L1 clone screening on Cyto-Chex spiked or not with rested PBMC.

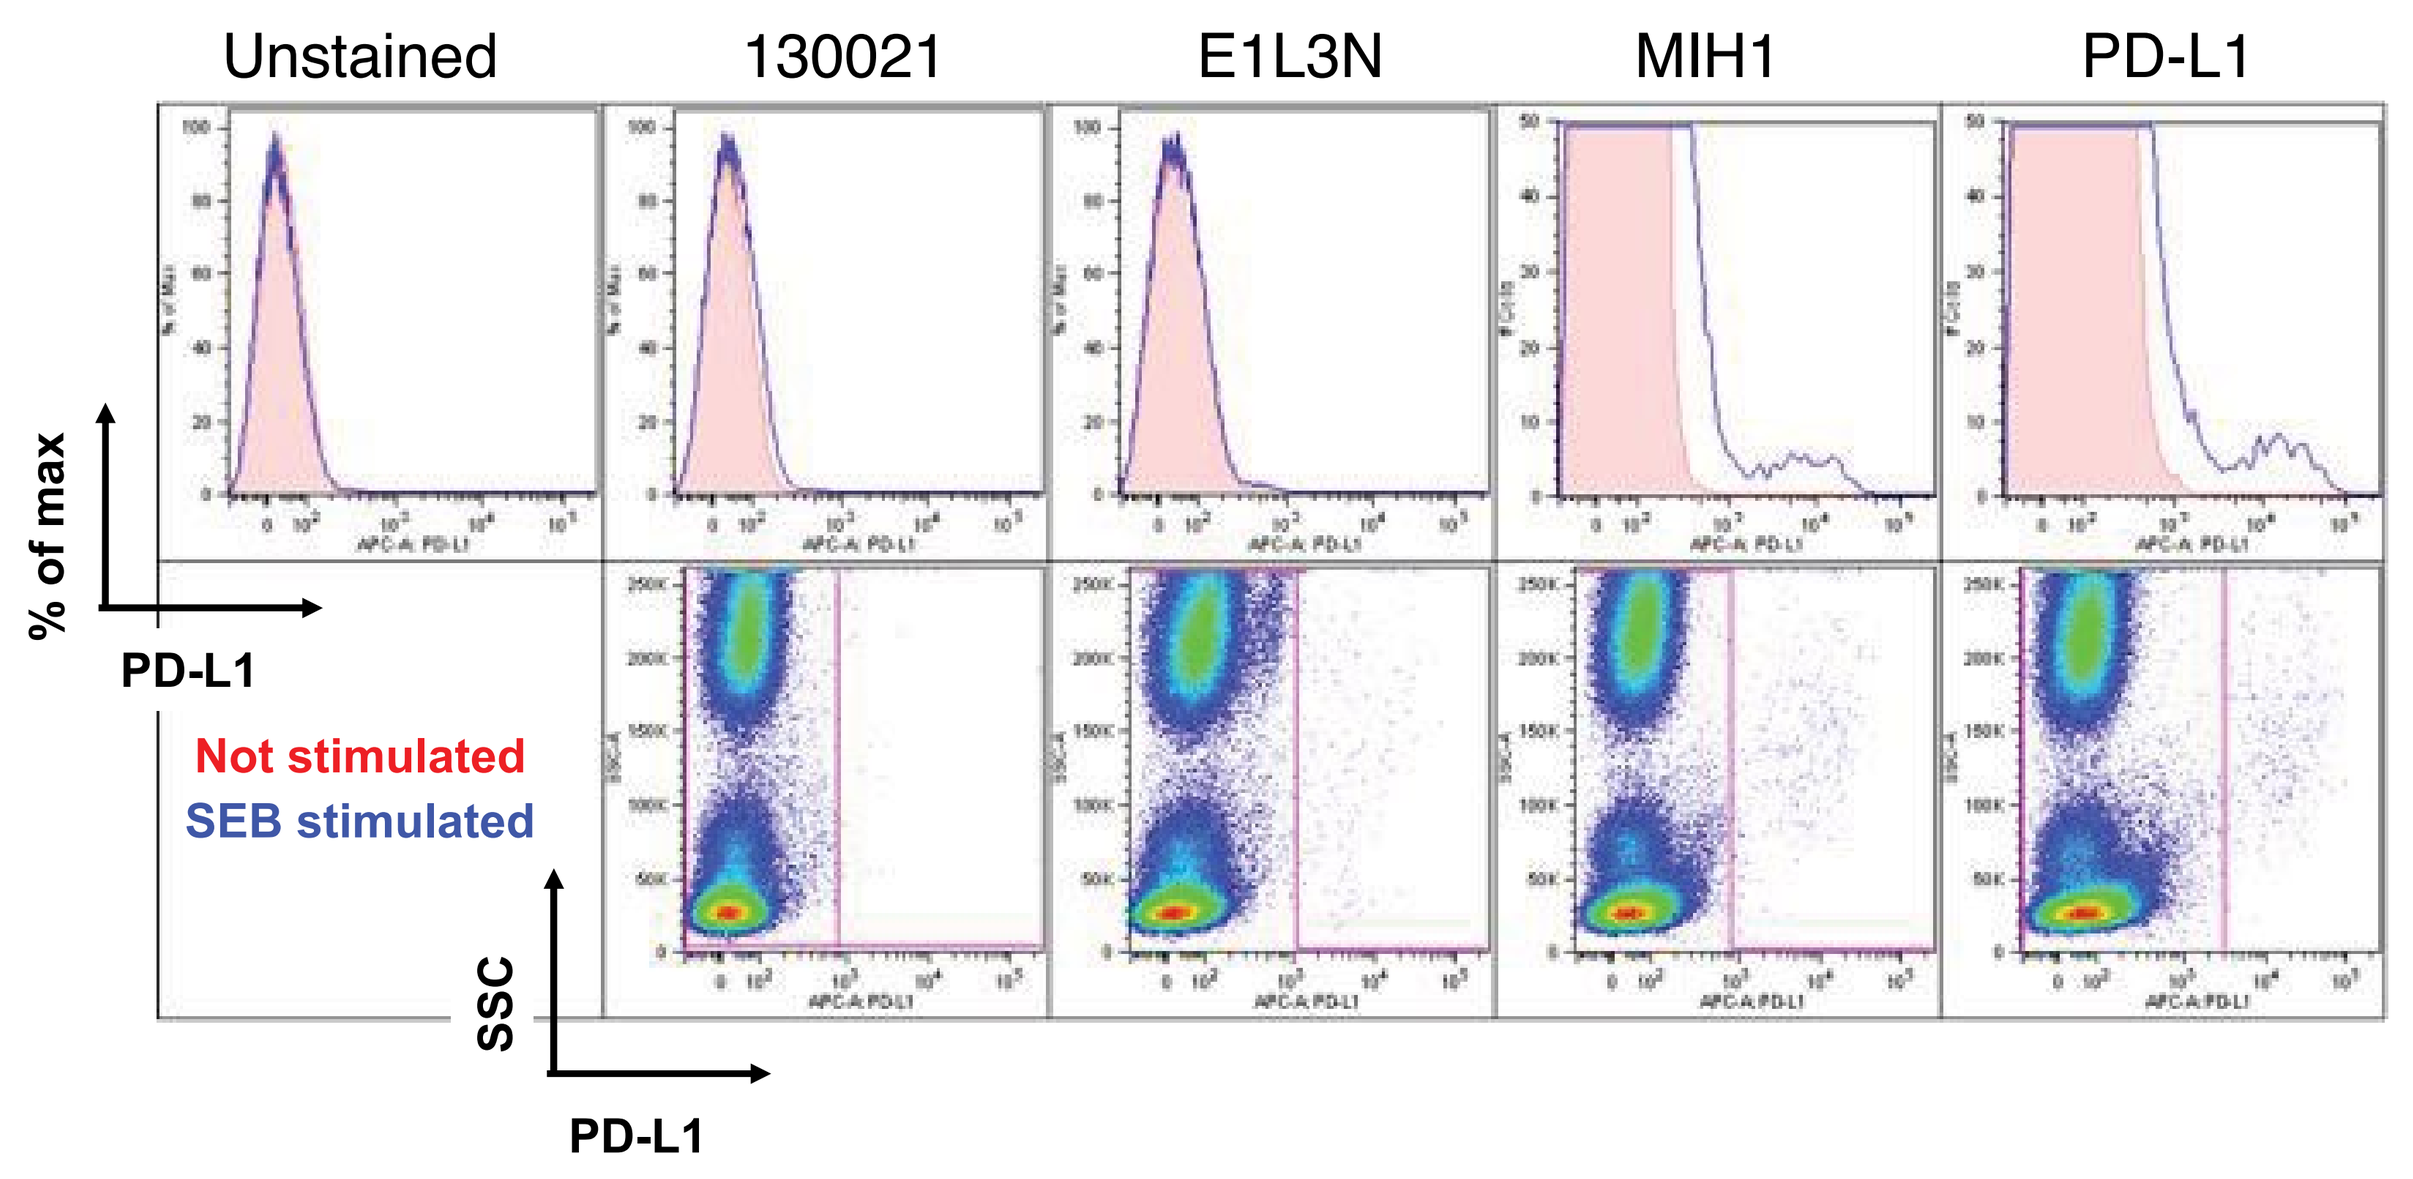
Unstained
130021
E1L3N
MIH1
PD-L1
% of max
PD-L1
Not stimulated
SEB stimulated
SSC
PD-L1

CONCLUSIONS

PD-1

- **EH12.2H7, EH12.1** and **eBioJ105** clones have a better staining resolution and lead to PD-1 staining with a large range of staining.
- **MIH4** (from BD Biosciences and Biolegend) has similar staining profile, with PD-1 positive staining in stimulated cells, but cannot detect PD-1 expression at basal level.

LAG-3

- All five clones screened in this work were able to detect LAG-3 induction in activated PBMC spiked in whole blood collected in CytoChex.
- The resolution of detection varied between the clones and identified between 13-25% of lymphocytes expressing LAG-3.

PD-L1

- **MIH1** and **PD-L1** clones were able to detect PD-L1 on whole blood spiked with rested PBMC at low levels
- **130021** and **E1L3N** clones were not able to detect PD-L1 expression with the tested methodology.

CAPRION.COM