B171

COMPENSATION MATTERS, BUT HOW OFTEN?

Bader Yassine-Diab, Dominic Gagnon, Olivier Gingras, Sandrina Da Fonseca, Audrey Gagnon, Philippe Pouliot, and Virginia Litwin

CAPRION

BACKGROUND

The need for fluorescence spill over compensation is well understood by the flow cytometry community, as is the process for establishing the optimal compensation matrix. What is less well understood is how frequently compensation should be established; and whether an existing compensation matrix can be applied, and if so, for how long. Initially, the recommendations were that compensation should be performed daily, but with the newer instruments, there are suggestions that every two months is acceptable.

PURPOSE

The purpose of this study was to evaluate alternative approaches to setting compensation daily.

SIGNIFICANCE

Setting compensation less frequently would result in cost savings to the laboratory and possibly more reliable data.

EXPERIMENTAL DESIGN

Three processes for setting the compensation were evaluated:

- Daily Compensation Settings (DCS)
- Fixed Reference Compensation Matrix (FRCM)
- Average Compensation Matrix (ACM)

The differences in the reported data from two panels using each of the three compensation approaches were compared.

- Simple panel (TBNK)
 - Whole blood, Lyse/No Wash with cell enumeration beads
 - 6-color
 - Bimodal population distribution
- Complex panel (T cell Subsets)
 - Whole blood, Lyse/ Wash
 - 15-color
 - Bimodal and non-bimodal population distribution

Data Analysis

Daily Compensation Settings (DCS)	Fixed Reference Compensation Matrix (FRCM)	Average Compensation Matrix (ACM)
Compensation matrix established for each run	Compensation matrix established during the assay development /optimization	Compensation matrix is derived from the mean value of the first 10 runs
Pros/Cons		
• High labor and reagent costs • Impacted by lot-to-lot reagent changes • Best approach for tandem dyes	• Low labor and reagent costs • Based on a single run • Will not account for instrument daily variation • Will not account for tandem dye variation.	• Low labor and reagent costs • Based on multiple runs and thus accounts for instrument variations • Also accounts for experimental and reagent variation

SIMPLE PANEL (TBNK)

Data Analysis

Simple panel (TBNK)

18 runs evaluated over 22 months

All failed or flagged samples were eliminated from the analysis.

Only runs from the same cytometer were compared

FRCM and ACM were applied (post-acquisition)

No modification to the gating was applied

The % difference in the reportable results (population frequency) compared to the initial results obtained with the DCS was calculated.

Figure 1a:

WB panel	TBNK
Fluorophore	Marker
FITC	CD3
PerCP-Cy5.5	CD45
APC	CD19
APC-Cy7	CD8
PE	CD16/CD56
PE-Cy7	CD4

Figure 1b:

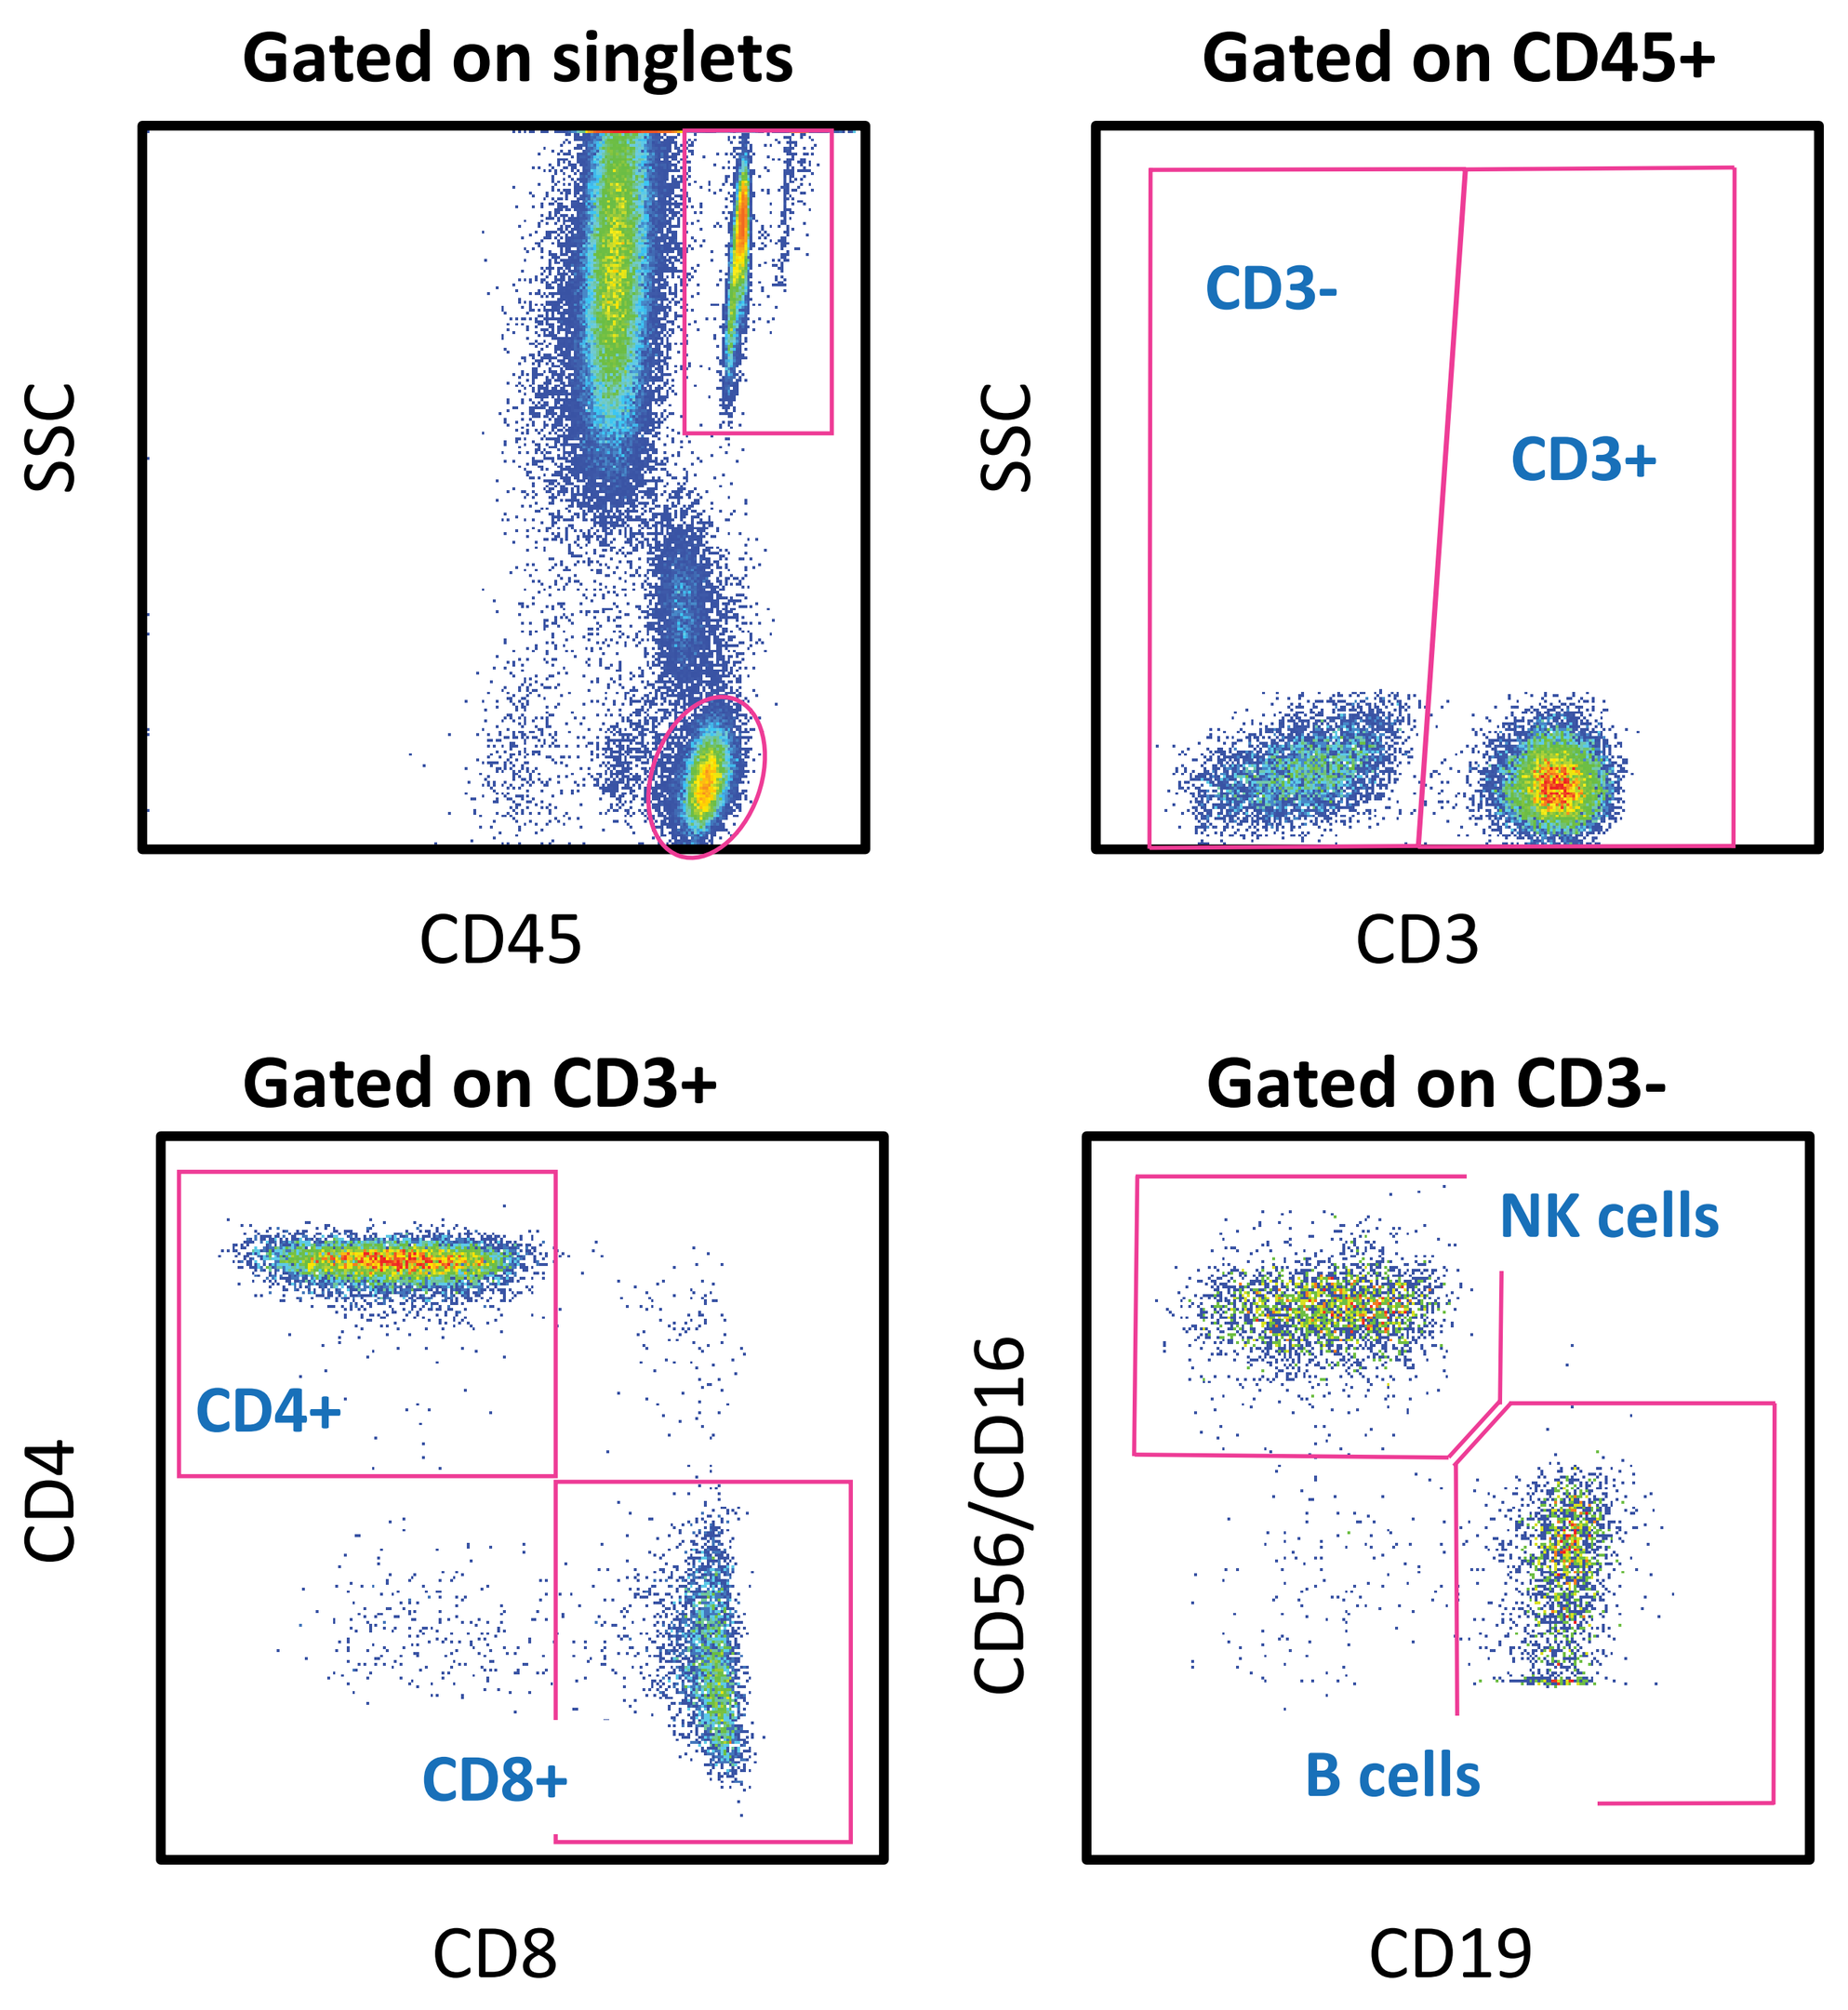
Gated on singlets
SSC
CD45
Gated on CD45+
CD3-
CD3+
SSC
CD3
Gated on CD3+
CD4+
CD8+
CD4
CD8
Gated on CD3-
NK cells
B cells
CD56/CD16
CD19

Figure 1c: Left Panel: The mean % difference in the reported value was over the 18 analytical runs was less than 1% when DCS was compared to FRCM (blue) or ACM (red).

Right Panel: Individual values over the 18 analytical runs.

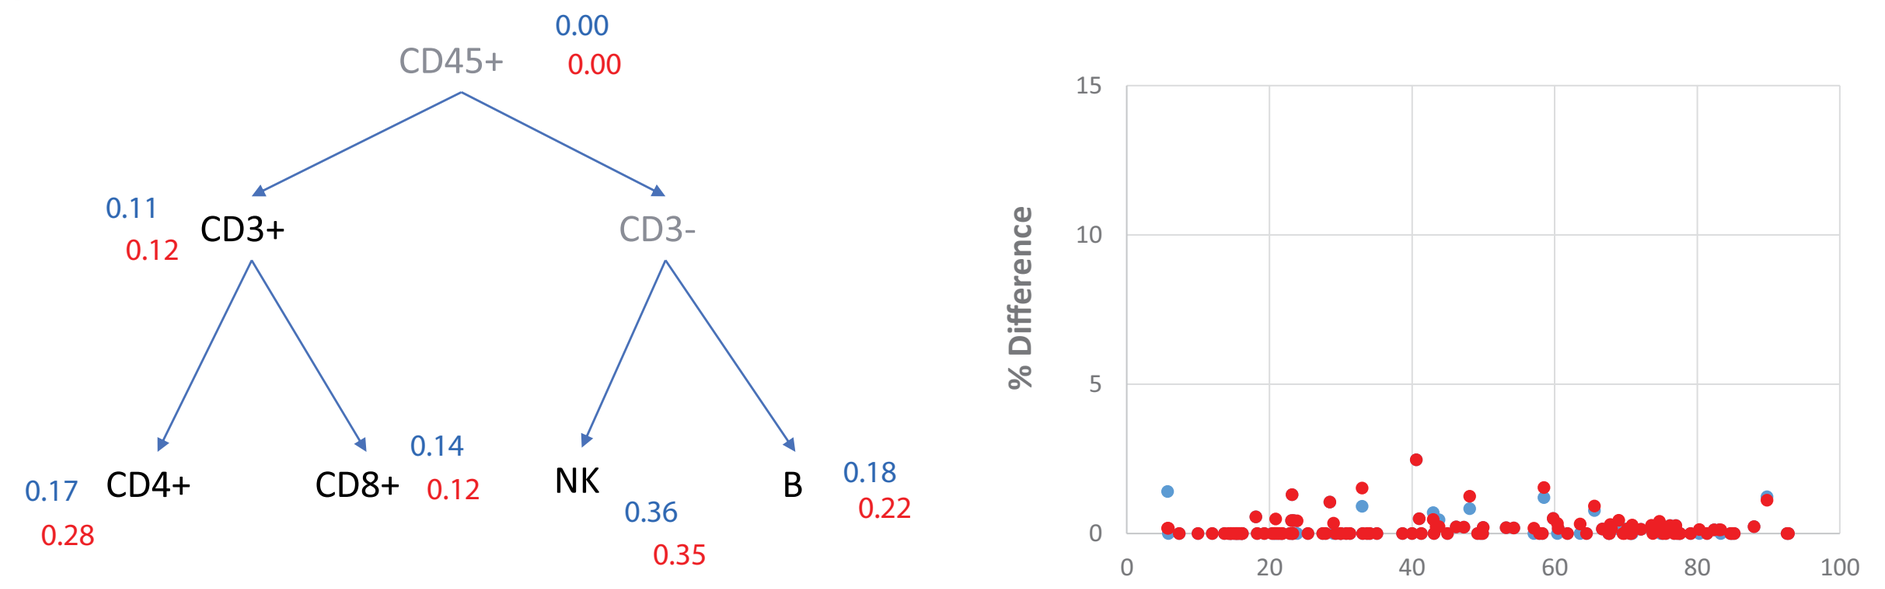
CD45+ 0.00 0.00
CD3+ 0.11 0.12
CD3-
CD4+ 0.17 0.28
CD8+ 0.14 0.12
NK 0.36 0.35
B 0.18 0.22
% Difference
Relative Percentage (from DCM)

Figure 1d: Longitudinal comparison of the % difference frequencies between DCS and FRCM (blue) and ACM (red) matrices from T cells and NK cells.

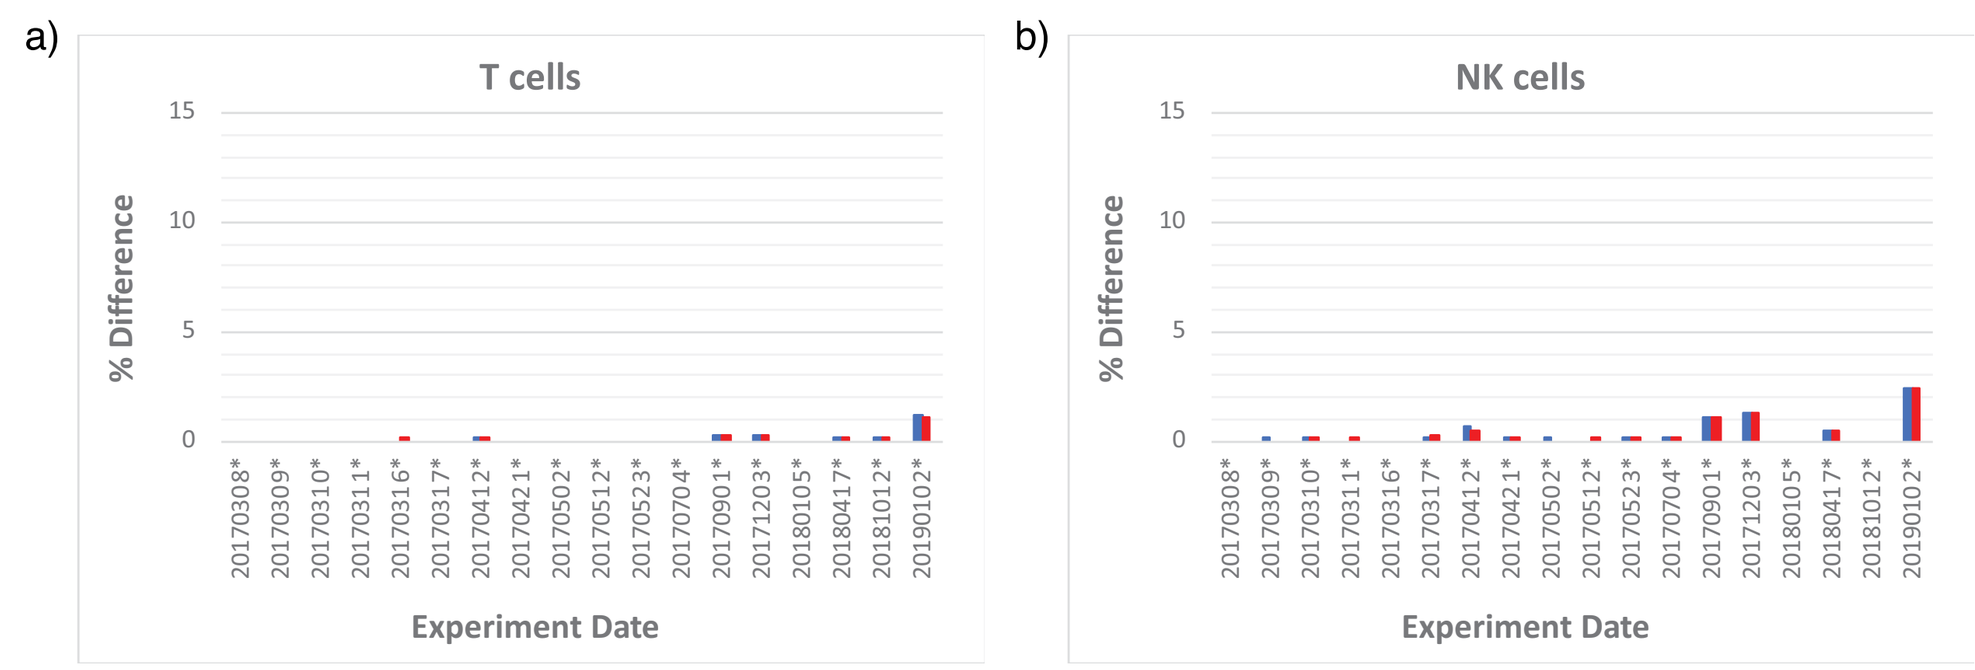
a)
T cells
b)
NK cells
% Difference
Experiment Date

COMPLEX PANEL (T CELL SUBSETS)

Figure 2a:

WB panel	T cell Subsets
Fluorophore	Marker
FITC	CD45
PerCP-Cy5.5	CD27
APC	KLRG-1
BV421	CD69
BV510	CD3
BV605	CCR7
BV650	ICOS
BV711	CD45RA
BV786	HLA-DR
PE	PD-1
PE-CF594	CD127
PE-Cy7	CD28
BV737	CD8
BV496	CD4
BV395	CD25

Complex panel (T cell Subsets)

6 runs evaluated over 7 months

All failed or flagged samples were eliminated from the analysis

Results with <30 events were eliminated from the analysis

Only runs from the same cytometer were compared

FRCM and ACM were applied (post-acquisition)

No modification to the gating was applied

The % difference in the reportable results (population frequency) compared to the initial results obtained with the DCS was calculated.

Figure 2b

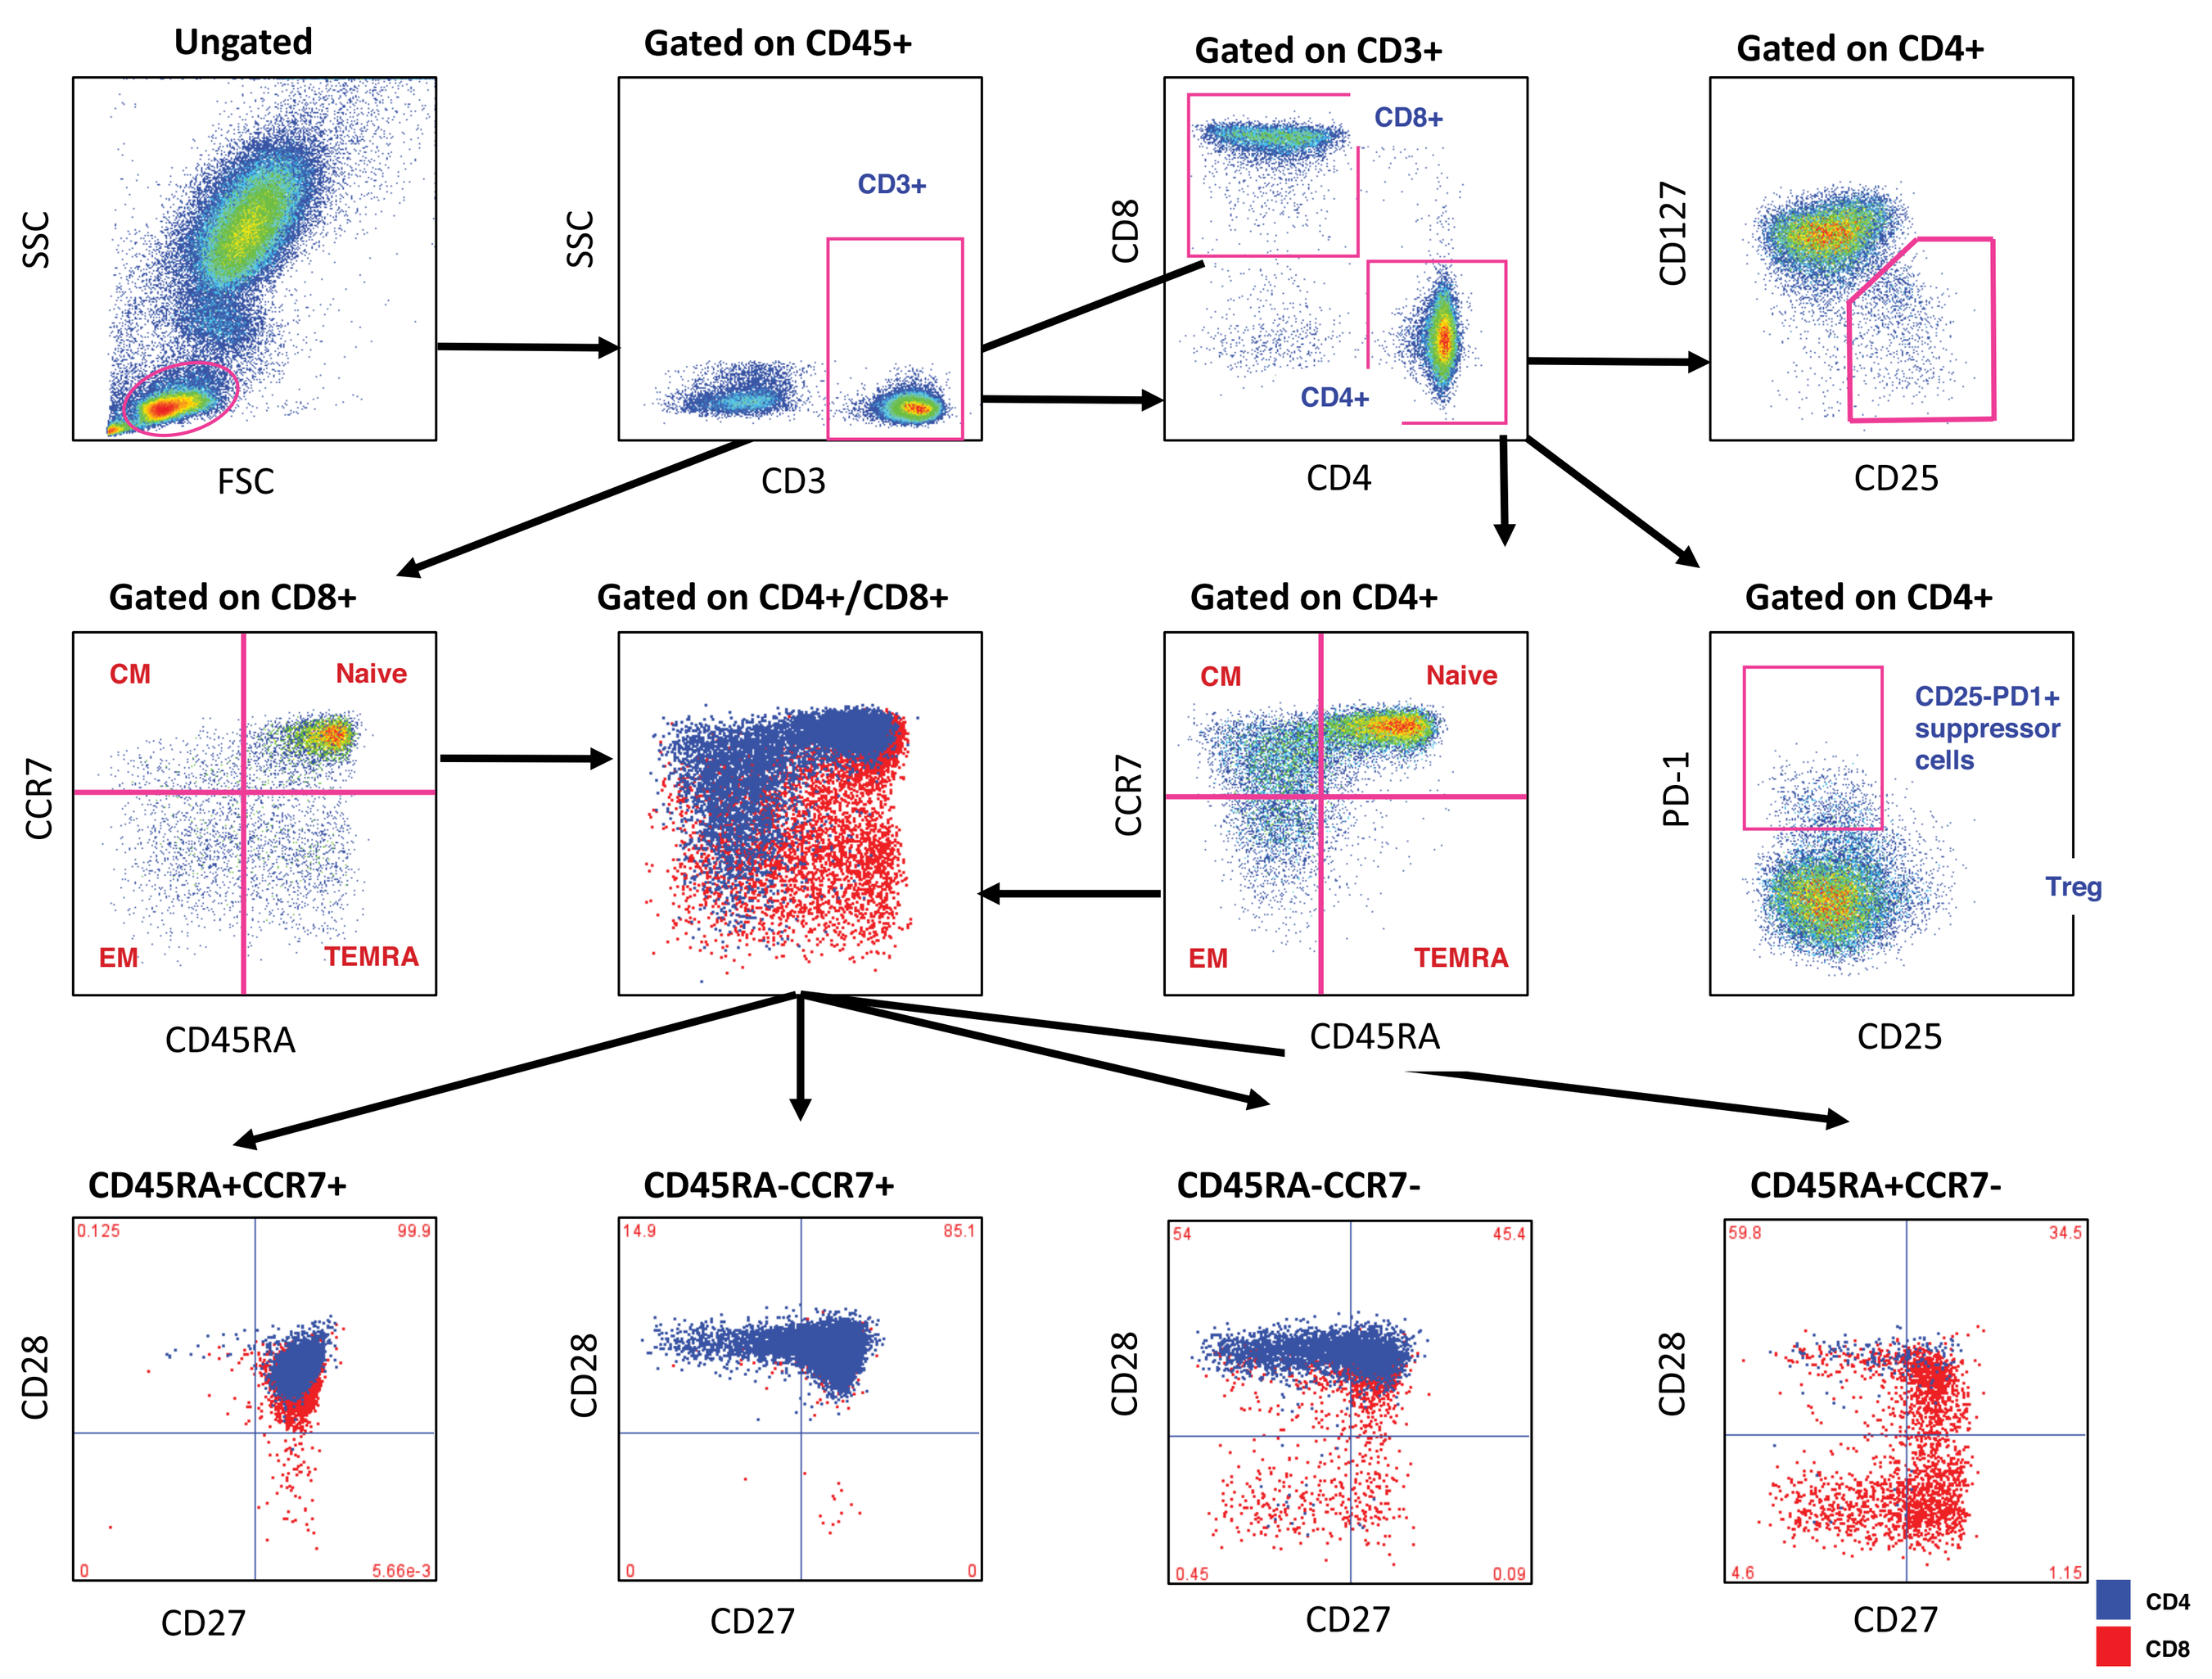
Ungated
SSC
FSC
Gated on CD45+
CD3+
SSC
CD3
Gated on CD3+
CD8+
CD4+
CD8
CD4
Gated on CD4+
CD127
CD25
Gated on CD8+
CM
Naive
EM
TEMRA
CCR7
CD45RA
Gated on CD4+/CD8+
Gated on CD4+
CM
Naive
EM
TEMRA
CCR7
CD45RA
Gated on CD4+
CD25-PD1+ suppressor cells
Treg
PD-1
CD25
CD45RA+CCR7+
CD45RA-CCR7+
CD45RA-CCR7-
CD45RA+CCR7-
CD28
CD27
CD4
CD8

Figure 2c

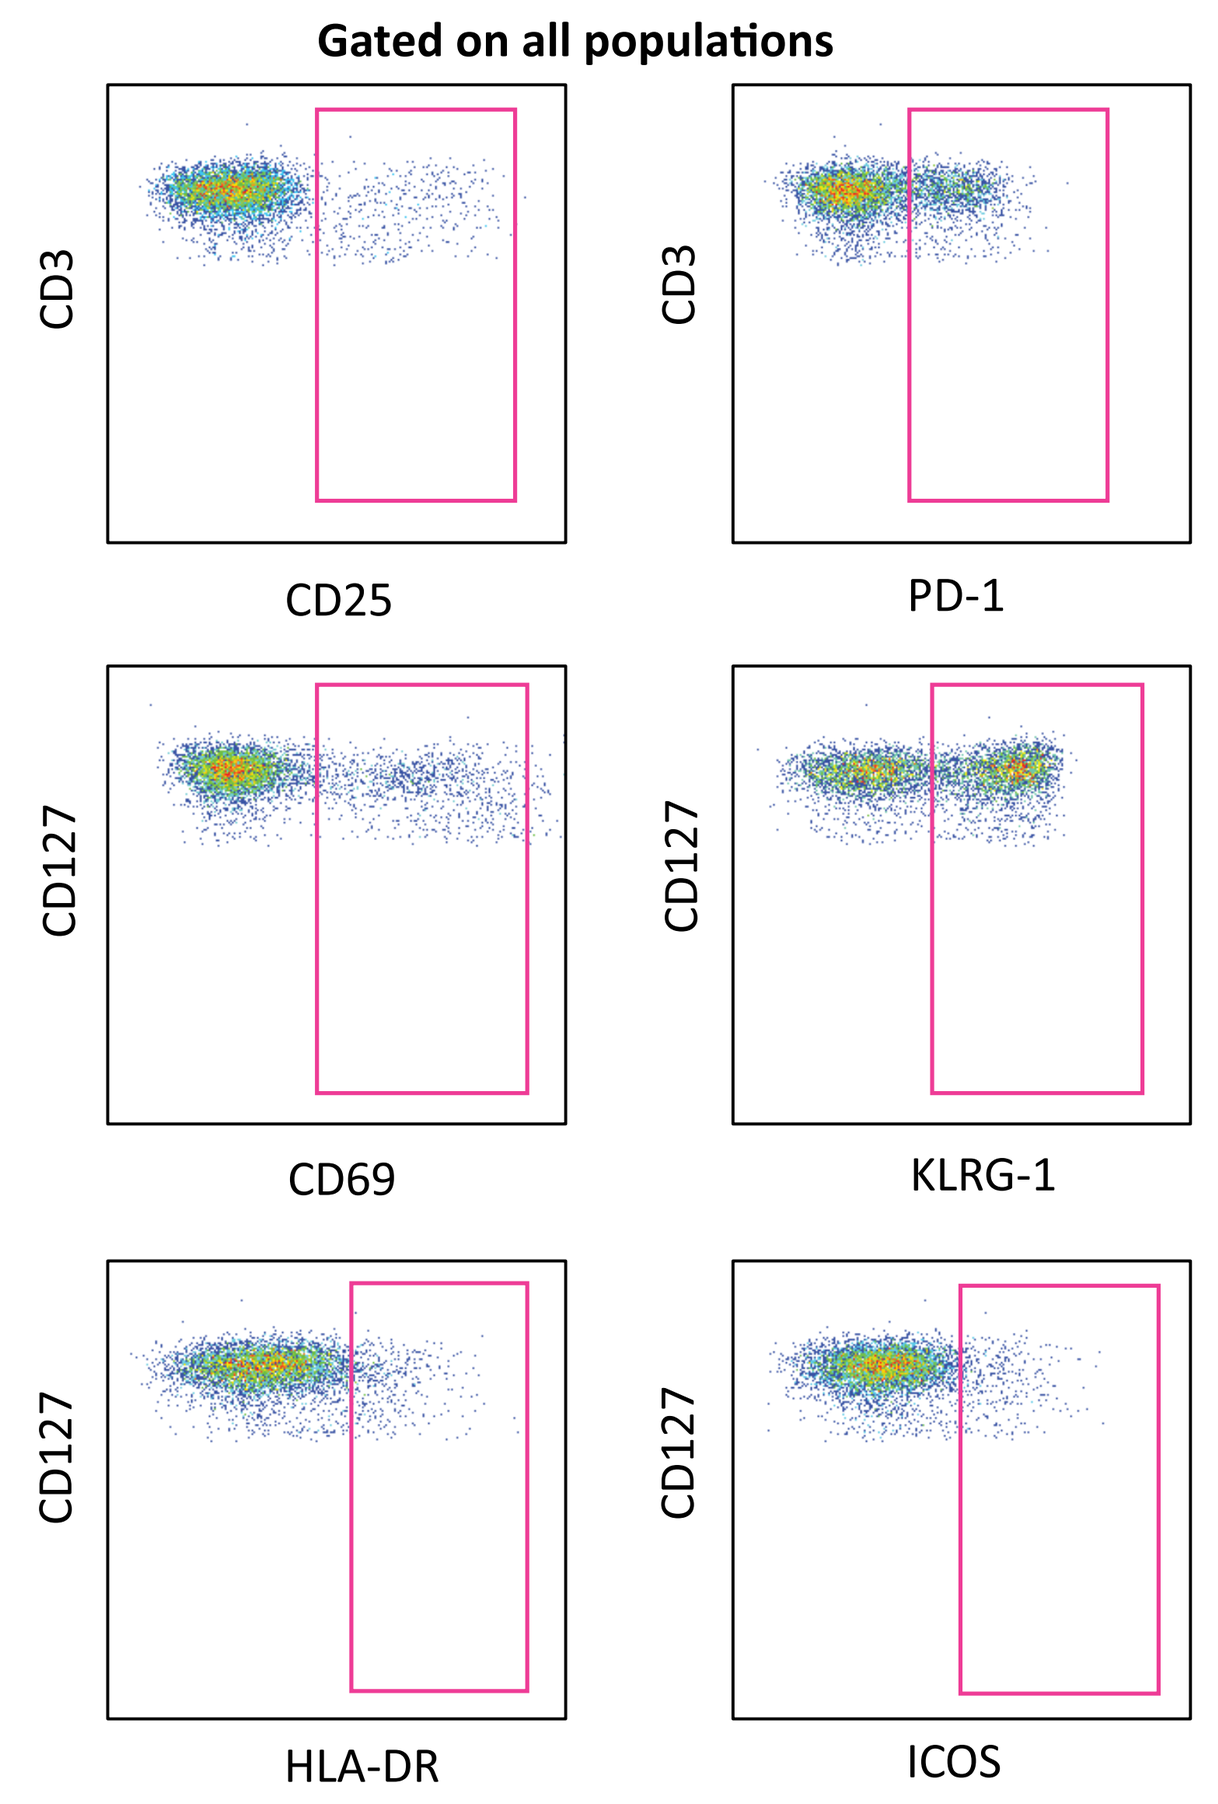
Gated on all populations
CD3
CD25
CD3
PD-1
CD127
CD69
CD127
KLRG-1
CD127
HLA-DR
CD127
ICOS

Figure 2d: Daily Compensation Settings

- Overlay of 10 runs over 4 weeks

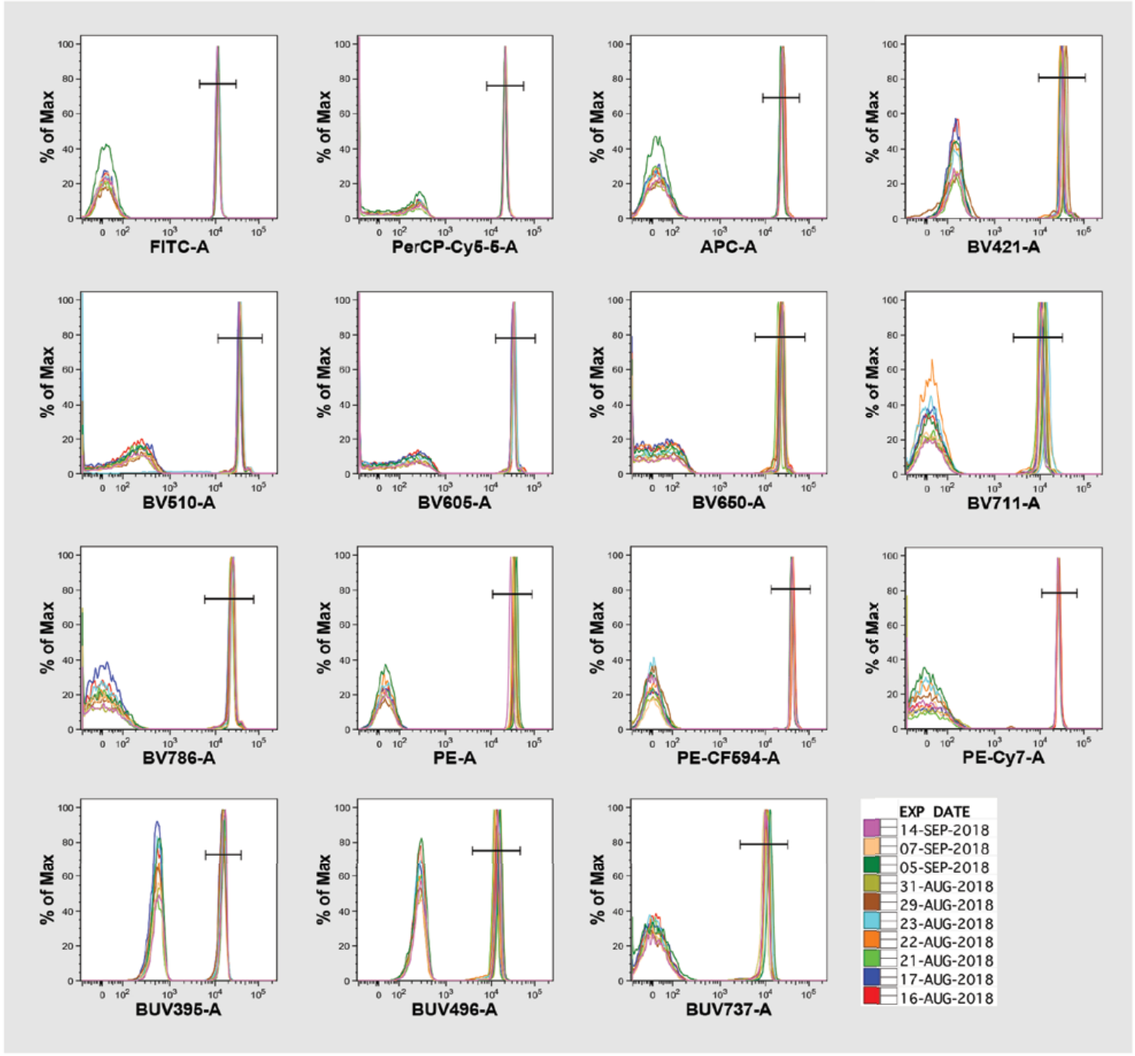

Figure 2e: Daily Compensation Settings

- Mean (top) and %CV (bottom) of 10 runs over 4 weeks

Average	FITC	PerCP-Cy5-5	APC	BV421	BV510	BV605	BV650	BV711	BV786	PE	PE-CF594	PE-Cy7	BUV395	BUV496	BUV737
FITC		2.05%	0.00%	0.00%	4.81%	0.67%	0.16%	0.06%	0.00%	0.00%	0.01%	0.00%	0.02%	5.98%	0.00%
PerCP-Cy5-5	0.00%		5.63%	0.00%	0.00%	0.01%	4.70%	34.58%	9.29%	0.00%	0.00%	3.46%	0.00%	0.00%	6.04%
APC	0.00%	1.19%		0.00%	0.00%	0.07%	9.25%	3.61%	0.72%	0.00%	0.10%	1.67%	0.01%	0.00%	2.78%
BV421	0.00%	0.00%	0.00%		17.94%	0.99%	0.22%	0.10%	0.00%	0.00%	0.00%	0.00%	0.00%	4.99%	0.00%
BV510	2.49%	0.06%	0.01%	18.74%		13.89%	3.63%	1.39%	0.24%	0.00%	0.00%	0.00%	0.00%	23.62%	0.12%
BV605	0.00%	2.11%	0.06%	0.52%	0.28%		45.96%	22.54%	6.10%	4.66%	11.22%	0.78%	0.00%	0.08%	2.81%
BV650	0.00%	1.20%	7.12%	2.29%	2.59%	18.89%		47.57%	9.71%	0.00%	0.44%	0.33%	0.00%	0.26%	4.50%
BV711	0.00%	10.18%	1.04%	3.16%	1.03%	0.23%	2.15%		46.21%	0.00%	0.00%	0.57%	0.04%	0.25%	20.12%
BV786	0.00%	0.02%	0.07%	2.89%	0.78%	0.24%	0.25%	3.46%		0.00%	0.00%	0.62%	0.00%	0.21%	3.95%
PE	0.95%	15.96%	0.01%	0.00%	0.23%	9.67%	2.45%	0.95%	0.14%		44.98%	0.95%	0.00%	0.00%	0.24%
PE-CF594	0.12%	63.85%	0.18%	0.00%	0.00%	18.25%	6.07%	3.70%	0.63%	15.92%		5.03%	0.00%	0.00%	0.99%
PE-Cy7	0.19%	0.50%	0.00%	0.00%	0.00%	0.11%	0.01%	0.26%	12.16%	1.27%	0.63%		0.00%	0.00%	1.60%
BUV395	0.00%	0.00%	0.00%	0.18%	0.01%	0.02%	0.00%	0.00%	0.00%	0.00%	0.00%	0.00%		14.45%	0.00%
BUV496	1.77%	0.04%	0.03%	0.53%	11.29%	1.13%	0.28%	0.13%	0.00%	0.00%	0.09%	0.00%	4.59%		0.37%
BUV737	0.00%	3.27%	0.15%	0.00%	0.00%	0.06%	0.03%	4.08%	4.79%	0.00%	0.00%	0.72%	4.98%	0.70%	

CV	FITC	PerCP-Cy5-5	APC	BV421	BV510	BV605	BV650	BV711	BV786	PE	PE-CF594	PE-Cy7	BUV395	BUV496	BUV737
FITC	0.00	4.45	0.00	0.00	2.07	11.29	0.00	0.00	0.00	0.00	0.00	0.00	0.00	2.69	0.00
PerCP-Cy5-5	0.00	0.00	2.94	0.00	0.00	0.00	1.89	2.46	2.45	0.00	0.00	2.50	0.00	0.00	3.22
APC	0.00	4.87	0.00	0.00	0.00	0.00	2.58	3.14	3.61	0.00	0.00	1.75	0.00	0.00	3.43
BV421	0.00	0.00	0.00	0.00	1.41	4.05	0.00	0.00	0.00	0.00	0.00	0.00	0.00	3.42	0.00
BV510	6.72	0.00	0.00	2.51	0.00	0.79	1.41	1.32	0.00	0.00	0.00	0.00	0.00	3.59	0.00
BV605	0.00	3.22	0.00	2.46	0.00	0.00	1.37	1.38	2.34	4.09	3.71	3.76	0.00	0.00	4.76
BV650	0.00	8.89	8.25	4.04	4.31	1.50	0.00	1.43	2.12	0.00	0.00	0.00	0.00	0.00	4.21
BV711	0.00	6.16	5.97	5.34	4.15	0.00	2.93	0.00	1.24	0.00	0.00	10.18	0.00	0.00	4.39
BV786	0.00	0.00	0.00	4.33	7.28	0.00	0.00	1.35	0.00	0.00	0.00	4.53	0.00	0.00	4.44
PE	3.14	3.03	0.00	0.00	0.00	4.09	3.88	4.40	0.00	0.00	0.97	9.30	0.00	0.00	0.00
PE-CF594	0.00	2.77	0.00	0.00	0.00	3.62	3.18	4.09	4.47	0.99	0.00	0.83	0.00	0.00	2.06
PE-Cy7	0.00	0.00	0.00	0.00	0.00	0.00	0.00	0.00	3.89	1.37	1.39	0.00	0.00	0.00	3.00
BUV395	0.00	0.00	0.00	0.00	0.00	0.00	0.00	0.00	0.00	0.00	0.00	0.00	0.00	1.61	0.00
BUV496	4.01	0.00	0.00	15.81	5.98	9.55	0.00	0.00	0.00	0.00	0.00	0.00	0.90	0.00	0.00
BUV737	0.00	6.47	0.00	0.00	0.00	0.00	0.00	6.13	5.30	0.00	0.00	4.58	3.06	6.24	0.00

Figure 2f: % difference of frequencies between DCS and FRCM (blue) or ACM (red)

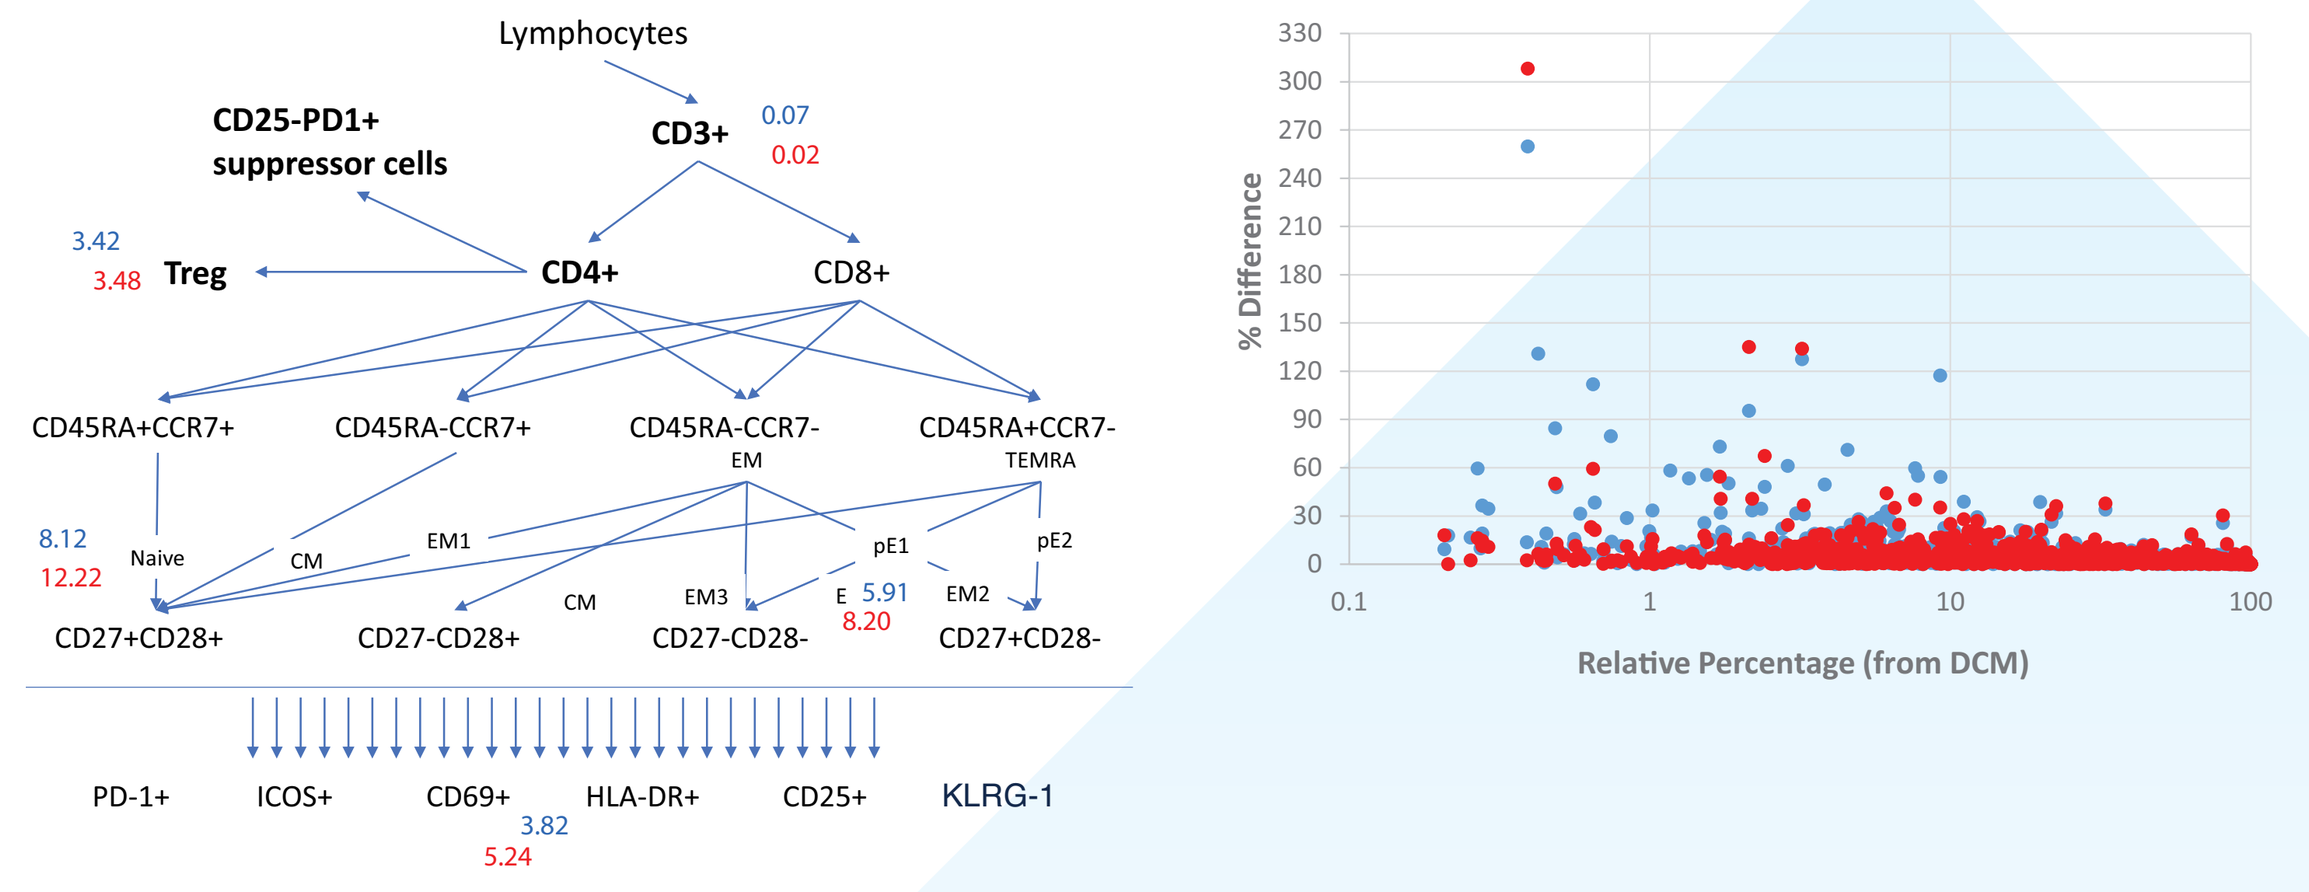
Lymphocytes
CD25-PD1+ suppressor cells
CD3+ 0.07 0.02
3.42 3.48 Treg
CD4+
CD8+
CD45RA+CCR7+
CD45RA-CCR7+
CD45RA-CCR7- EM
CD45RA+CCR7- TEMRA
8.12 12.22 Naive
CM
EM1
pE1
pE2
CM
EM3
E 5.91 8.20
EM2
CD27+CD28+
CD27-CD28+
CD27-CD28-
CD27+CD28-
PD-1+
ICOS+
CD69+ 3.82 5.24
HLA-DR+
CD25+
KLRG-1
% Difference
Relative Percentage (from DCM)

Conditions

- 893 results evaluated
- Gates were not changed

Results

- Comparison of DCS to FRCM (blue)
 - Overall higher percent differences were observed with FRCM (blue) compared to ACM (red)
- Comparison of DCS to ACM (red) (see pie chart next column)

Figure 2g: % difference between DCS and the AMC compensation matrices

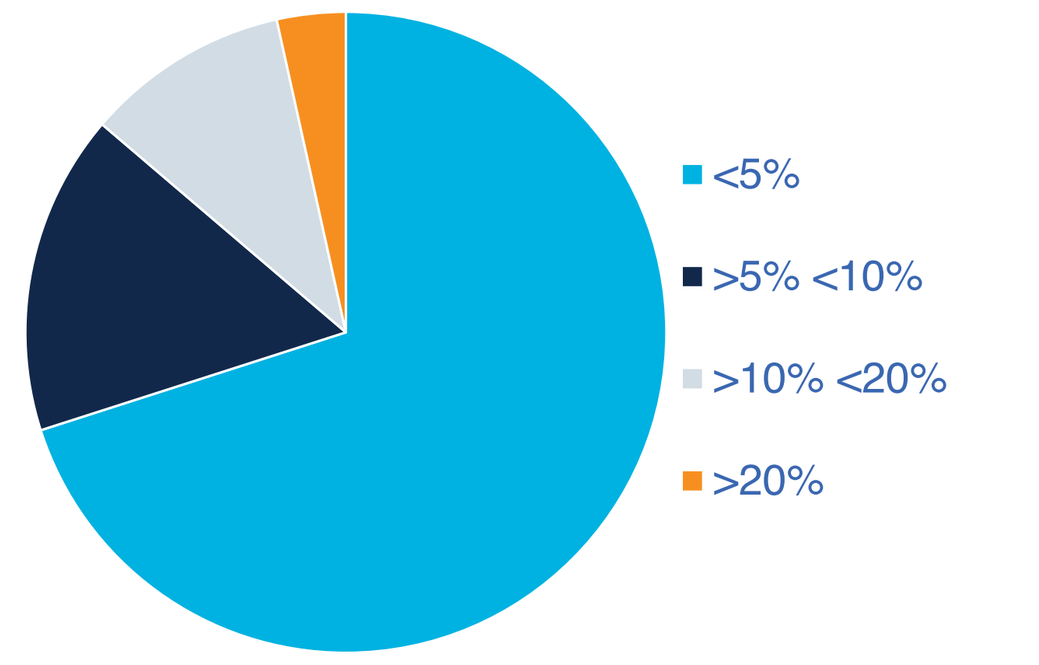
<5%
>5% <10%
>10% <20%
>20%

Conclusions

- Gate adjustments may be required after new compensation matrices are applied, as appropriate.

Next Steps

- See if gate adjustment improves correlation for ACM

Figure 2h: Changing of the gates after applying FRCM or ACM results in better correlation to original data with DCS

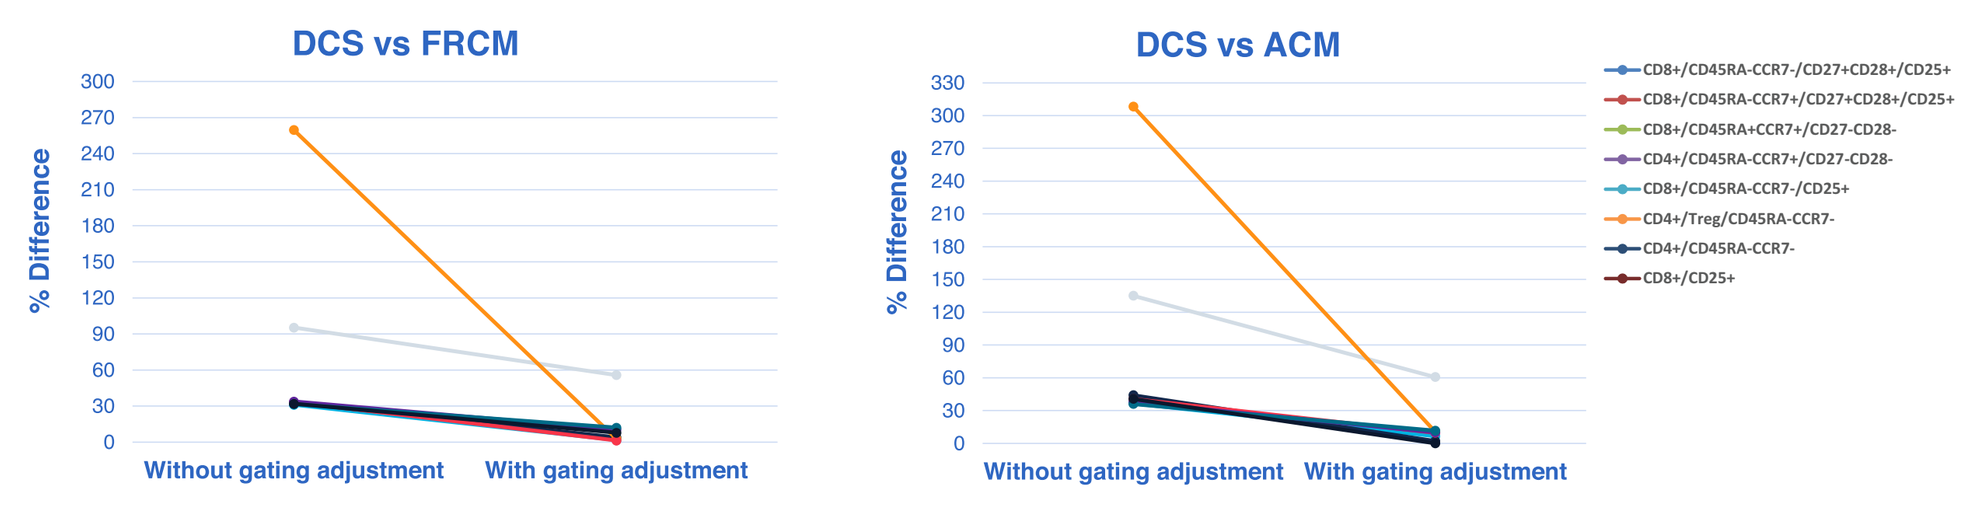
DCS vs FRCM
DCS vs ACM
% Difference
Without gating adjustment
With gating adjustment

CONCLUSIONS

"You don't know until you do the experiment." Georges Köhler

- In this study we evaluated three different processes for establishing the compensation matrix.
- For the Simple Panel (TBNK), where the reportable results are from bimodal population distributions, both the FRCM and the ACM provide consistent data to DCS. Performing compensation can result in cost savings.
- For the Complex Panel (T cell subsets), where the reportable results are from bimodal and non-bimodal population distributions, the ACM resulted in 68% of acceptable data (i.e. <5% difference) when no gates were changes. Overall the FRCM resulted higher variability compared to the ACM.
 - The results raised many additional, interesting questions.
 - Should the gates be changed with the change in compensation matrix?
 - The answer here is yes, see figure 2h.
 - Are some of the differences due to the fact that the initial data were sub-optimal?
 - Note that some of the differences observed with the changes in the compensation are also attributed to a minor software issue with FlowJo where the values from bi-exponential plots in a .jo file change slightly when the file is re-opened. This phenomenon is known as "picket fencing".

CAPRION.COM